Novel, brain penetrant small molecule inhibitor of PD-L1 for targeting glioblastoma and brain metastasis

Dhanalakshmi Sivanandhan[1], Sridharan Rajagopal[1], Chandru Gajendran[1], Naveen Sadhu M[1], Zainuddin Mohd[1], Ramachandraiah Gosu[1], **Luca Rastelli[2]**
[1]Jubilant Therapeutics India Limited, Bangalore, India. [2]Jubilant Therapeutics Bedminster, NJ.
Jubilant Therapeutics, Bedminster, NJ

SUMMARY

Background: The PD-1/PD-L1 molecular pathway is one of the primary mechanisms of immune evasion deployed by cancer cells. Activation of PD-1/PD-L1 pathway induces anergy and exhaustion of cytotoxic T-cells and enhances the function of regulatory T-cells causing an immune suppressive environment. Therefore, blocking this pathway restores T-cell proliferation and enhanced tumor cell killing. Approved monoclonal antibodies targeting PD-1/PD-L1 pathway require intravenous injections and have a long half-life that could contribute to the well-documented drug-related adverse effects. Also, efficacy of these antibodies appears to be marginal in malignancies associated with CNS due to poor brain penetrance. Therefore, small molecule inhibitors of the PD-1/PD-L1 pathway with oral bioavailability, better tumor and brain penetrance and shorter half-life will be highly valuable in cancer therapy. In this regard, JBI-2174 shows excellent ADME properties, brain exposure, pharmacokinetics and comparable efficacy as approved mAbs in preclinical studies.

Materials and methods: Structure based drug design was used to design PD-L1 inhibitors; potency of these inhibitors was assessed in an *in-vitro* TR-FRET assay. Reporter assays and ex-vivo co-culture assays were used to assess T-cell proliferation and function. Pharmacokinetics were performed in multiple pre-clinical species to derive at bioavailability and brain penetration. In vivo efficacy was assessed in partially humanized mice efficacy models.

Results: JBI-2174 showed strong *in vitro* IC_{50} of 1.5 nM in TR-FRET assay that measures interaction between PD-1 and PD-L1 and let to stabilization of PD-L1 as measured by thermal shift assay. This molecule also augmented T-cell co-inhibitory signalling as observed by Jurkat cell/SHP-1 assay. Competition study between anti-PD-L1 blocking antibody and x-ray crystal structure studies clearly demonstrated that JBI-2174 leads to dimerization of PD-L1. More importantly, JBI-2174 showed excellent oral bioavailability across pre-clinical species and strong and sustained (up to 24 h) brain exposure (0.66 to 2.1 fold plasma vs. brain ratio). JBI-2174 showed comparable or better efficacy to the anti-PD-L1 antibody Atezolizumab in hPD-L1/MC38 syngeneic and brain orthotopic models by oral administration with a concomitant increase in tumor infiltrating lymphocytes. Toxicological studies conducted in non-human primates clearly show that the molecule is well tolerated at exposures much higher than efficacious exposure.

Conclusion: Oral administration and brain exposure of these small molecule PD-L1 inhibitors provides an attractive option to be used in the treatment of glioblastoma and other solid tumors with brain metastasis. IND enabling studies are being initiated for this molecule to enable clinical trials in humans.

JBI-2174 is highly potent against PD-L1

a

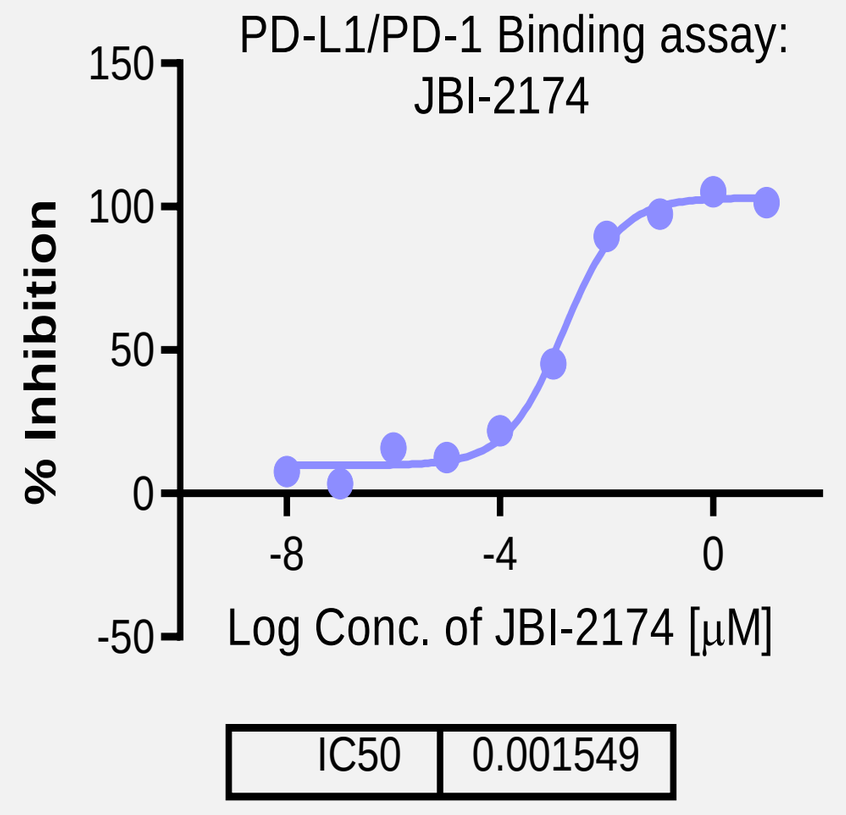
PD-L1/PD-1 Binding assay:
JBI-2174
% Inhibition
Log Conc. of JBI-2174 [μM]
IC50 0.001549

b

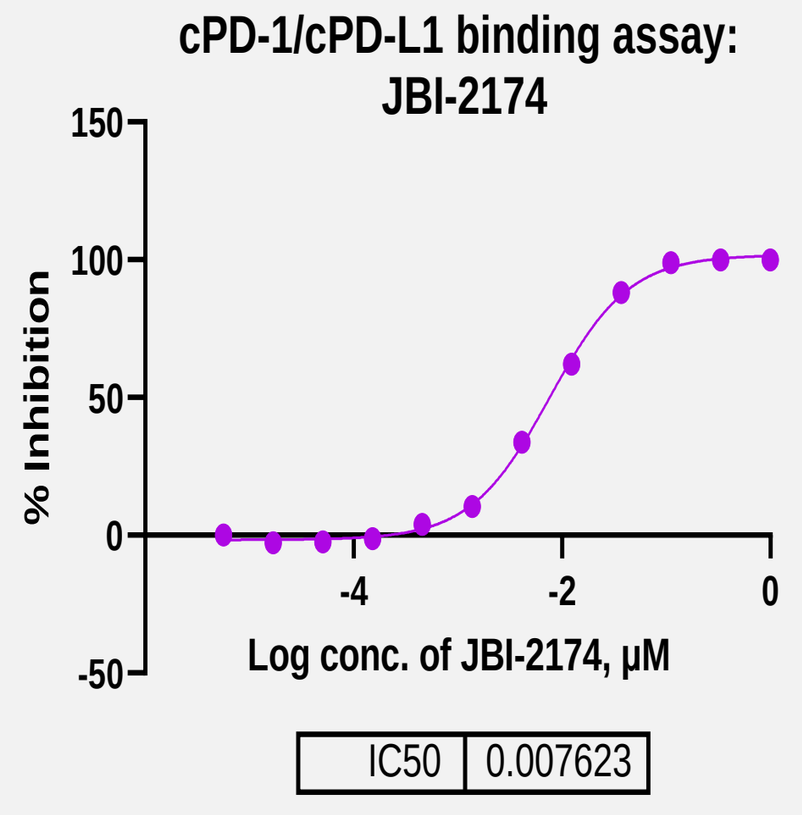
cPD-1/cPD-L1 binding assay:
JBI-2174
% Inhibition
Log conc. of JBI-2174, μM
IC50 0.007623

In vitro potency of JBI-2174 against (a) human PD-1/PD-L1 assessed by HTRF and (b) cyno PD-1/PD-L1 was assessed by ELISA based Binding Activity assay done at BPS Biosciences

JBI-2174 binds to PD-L1 leading to dimerization of PD-L1

a

PD-L1 dimerization by JBI-2174 (in-house structure at 2.6Å) | Footprints of PD-1 on PD-L1 | Footprints of JBI-2174 on PD-L1

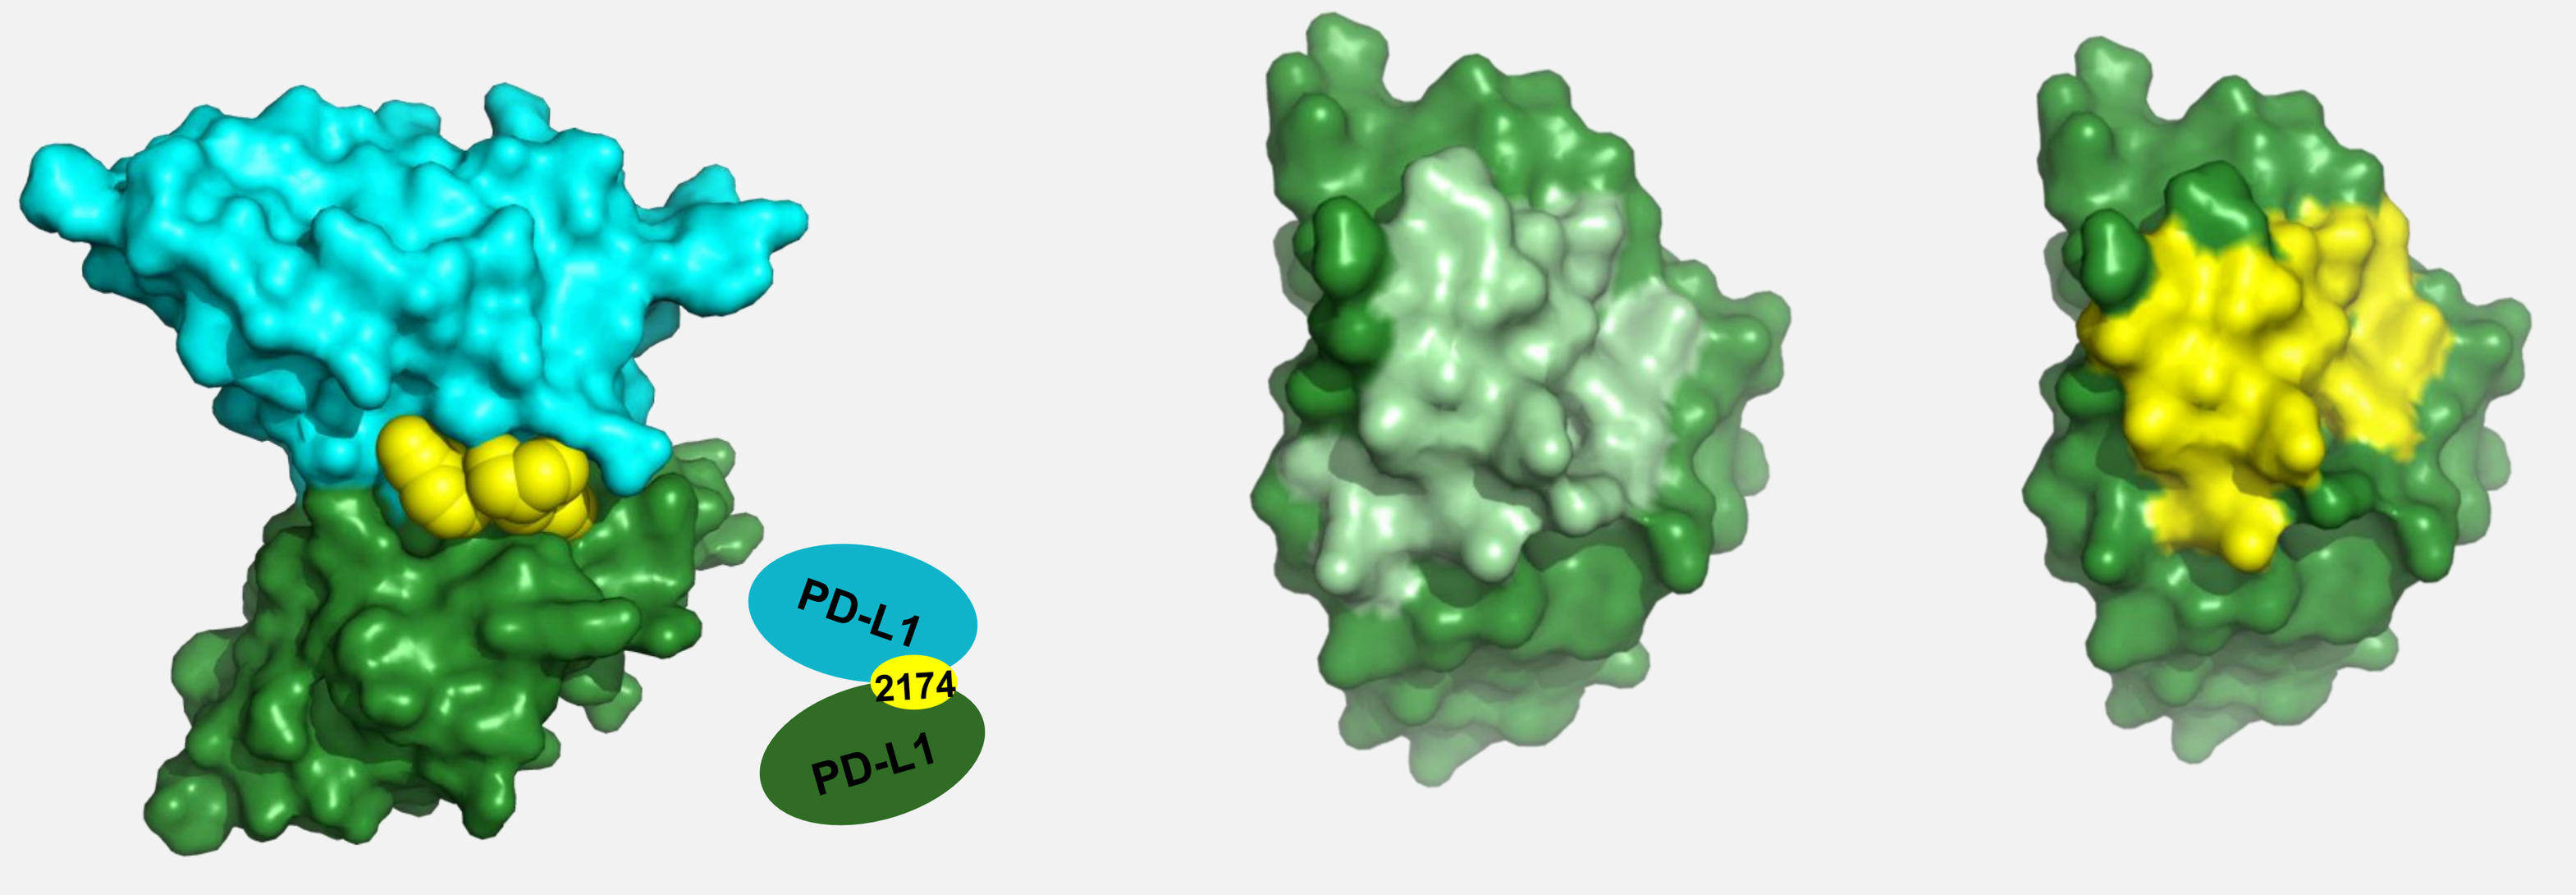
PD-L1
2174
PD-L1

- JBI-2174 binds selectively to PD-L1 and covers 65% of PD-1 footprint, similar to mAbs

JBI-2174 interrupts PD-1/PD-L1 signaling

PD-L1 dimerization: U2OS cells

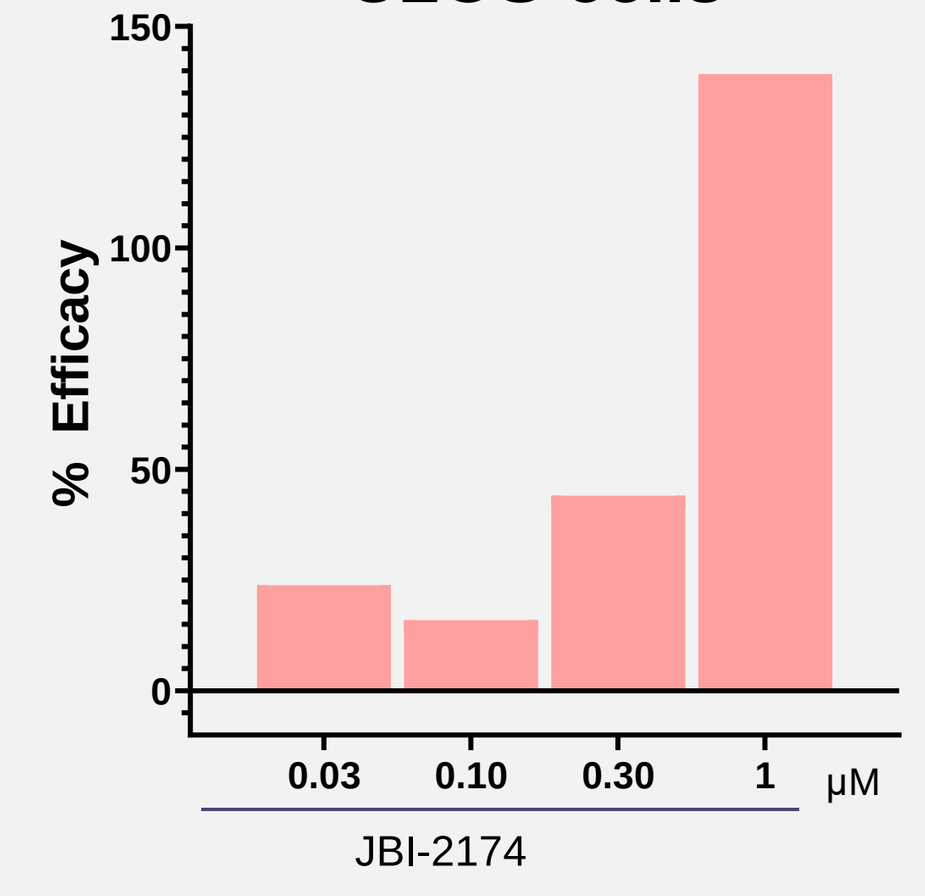
% Efficacy
0.03
0.10
0.30
1
μM
JBI-2174

PathHunter dimerization assay in U2OS cells(DiscoveRx)

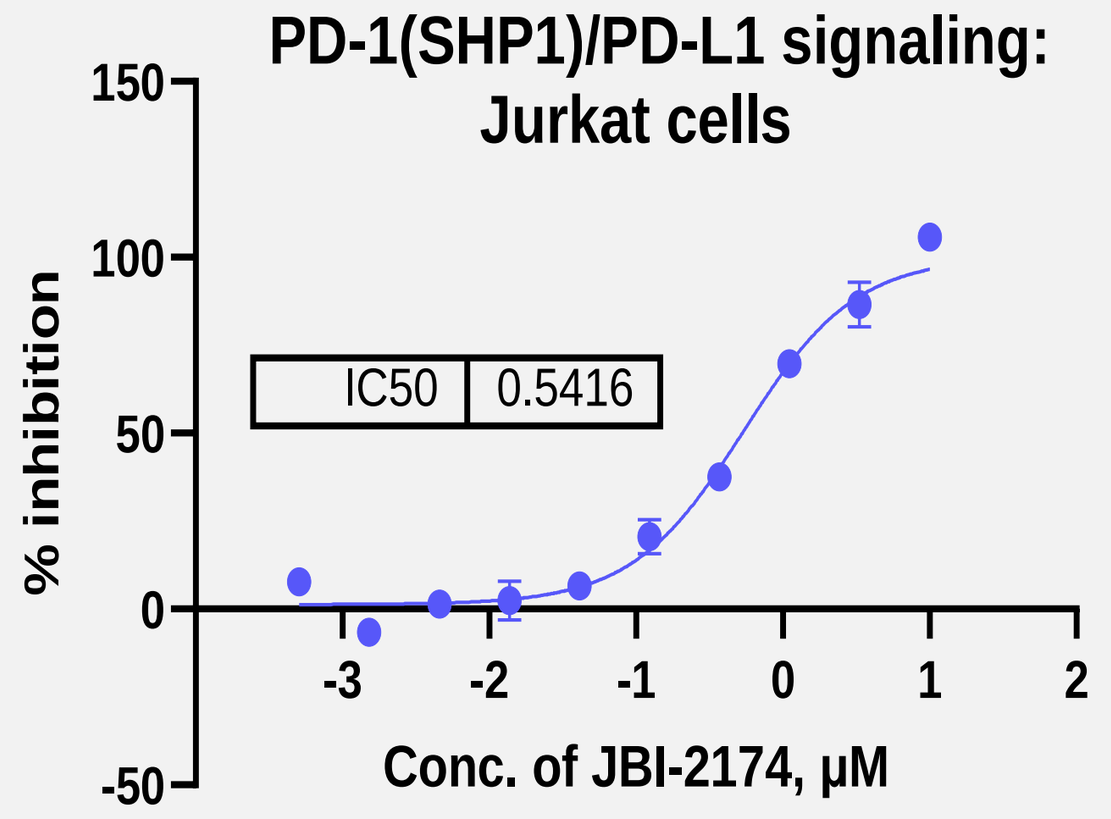
PD-1(SHP1)/PD-L1 signaling:
Jurkat cells
% inhibition
IC50 0.5416
Conc. of JBI-2174, μM

PathHunter PD-1 signaling assay in Jurkat cells (DiscoveRx)

- JBI-2174 leads to dimerization of PD-L1
- Inhibits PD-1/PD-L1 signaling

JBI-2174 is highly brain-penetrant

Exposure in SD rat

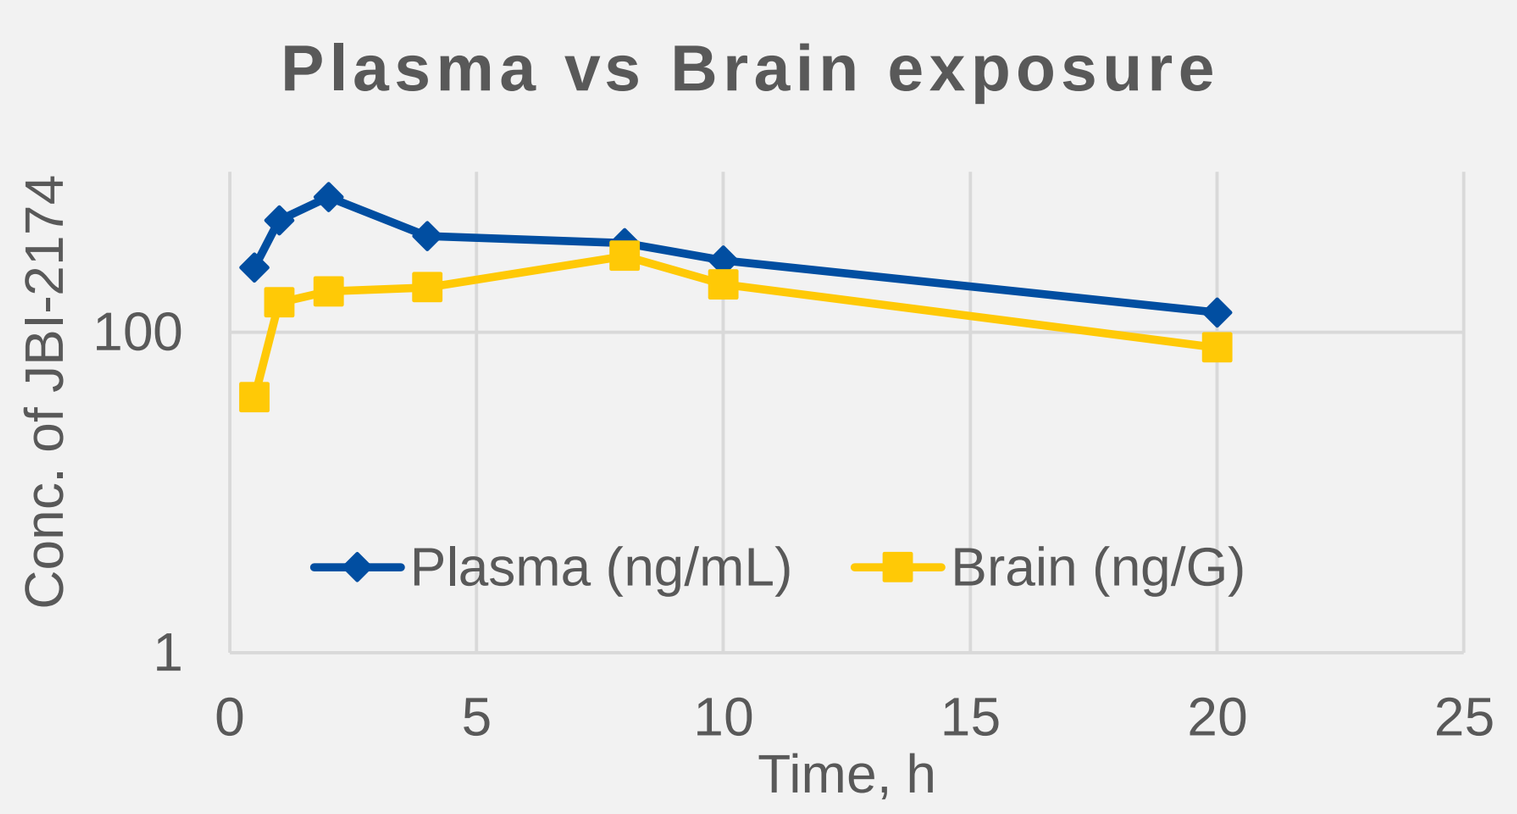
Plasma vs Brain exposure
Conc. of JBI-2174
Plasma (ng/mL)
Brain (ng/G)
Time, h

Exposure in mice

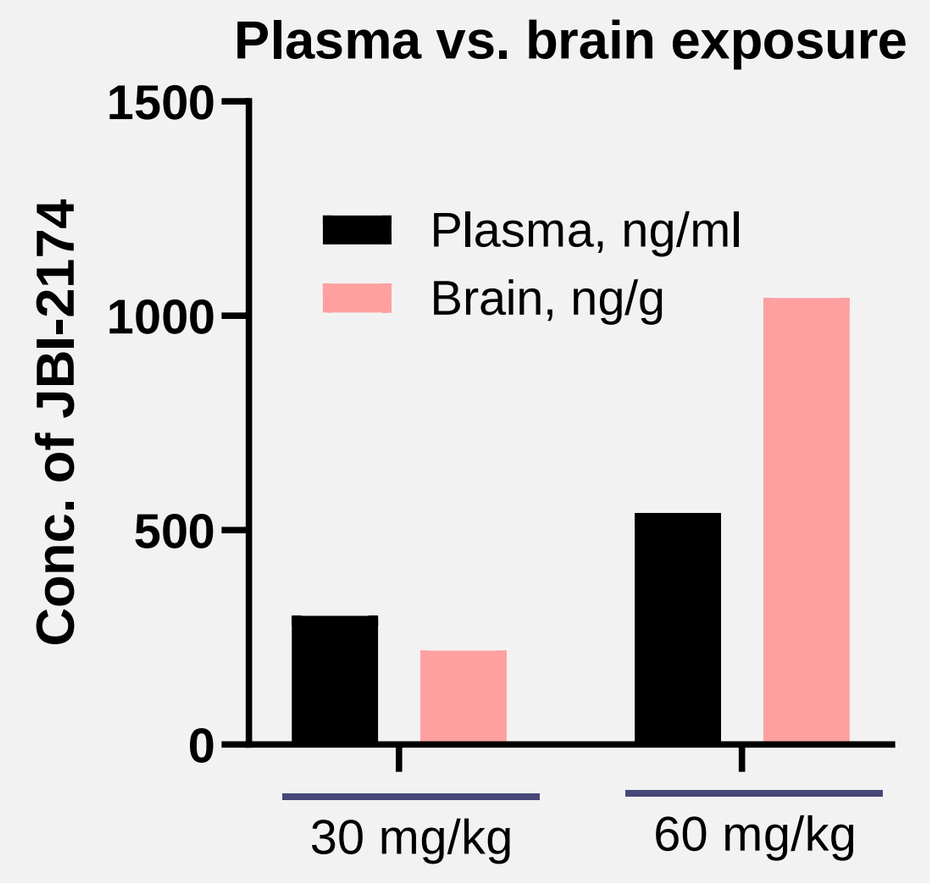
Plasma vs. brain exposure
Conc. of JBI-2174
Plasma, ng/ml
Brain, ng/g
30 mg/kg
60 mg/kg

Note: Conc. given are 24 h post dose

JBI-2174 extends survival in MC38/hPD-L1 orthotopic and syngeneic models

a

Brain orthotopic model: MC-38/hPD-L1

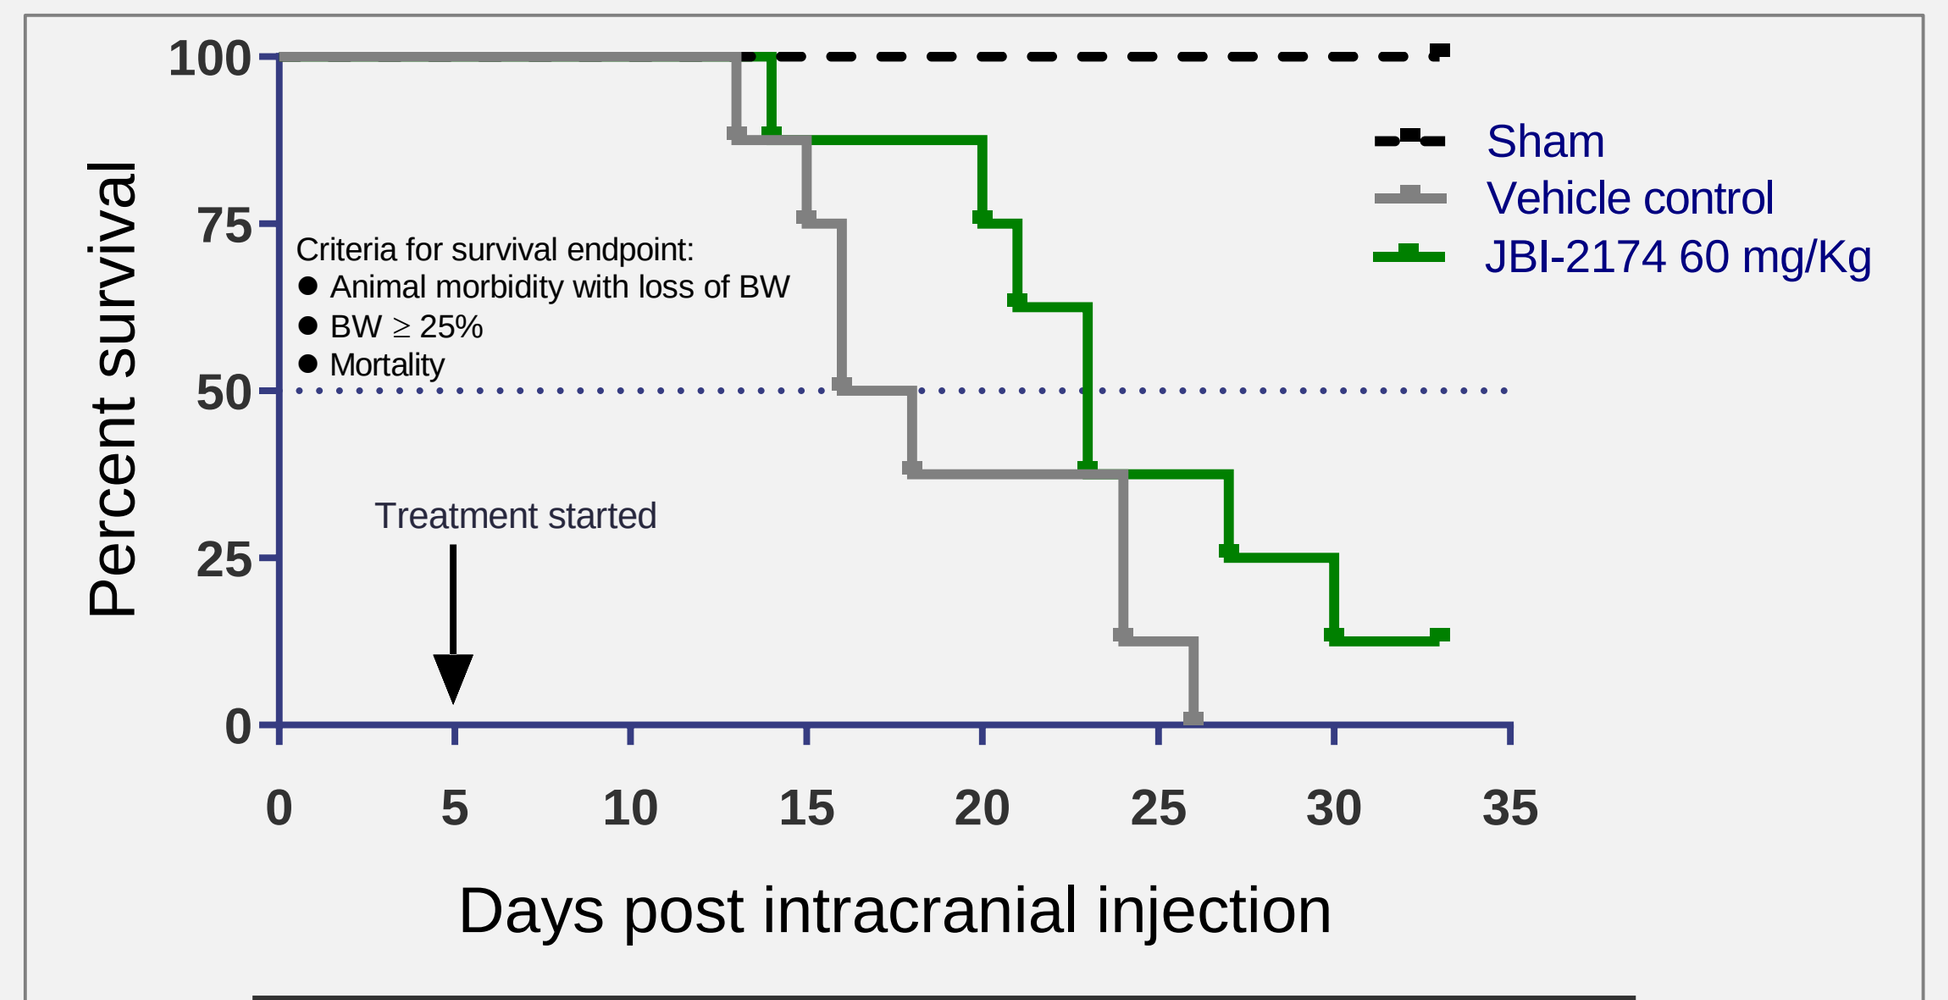
Percent survival
Criteria for survival endpoint:
Animal morbidity with loss of BW
BW ≥ 25%
Mortality
Treatment started
Sham
Vehicle control
JBI-2174 60 mg/Kg
Days post intracranial injection

	Sham	Vehicle control	JBI-2174, 60 mg/Kg, bid
Median survival	Undefined	17	23

b

Syngeneic model: MC-38/hPD-L1

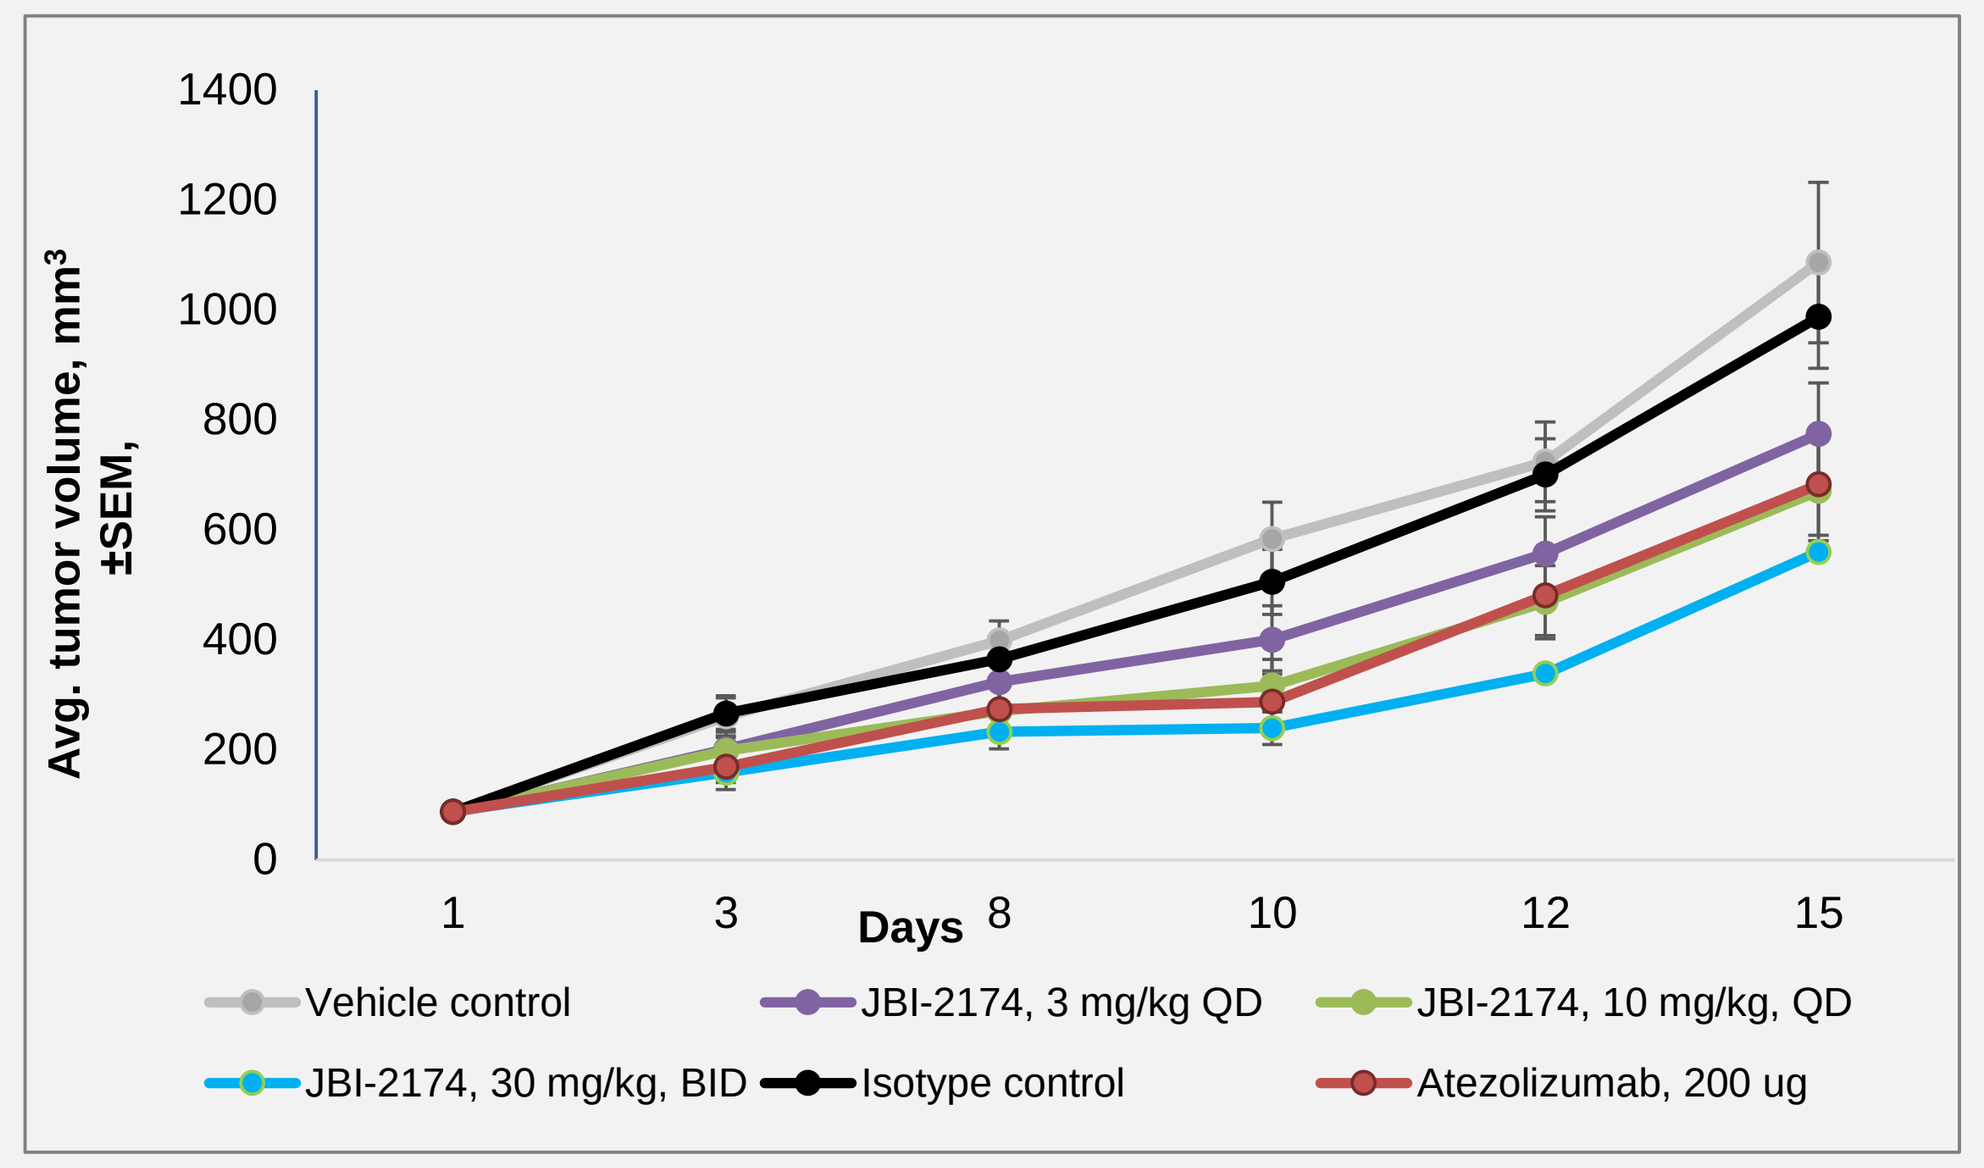
Avg. tumor volume, mm³ ±SEM,
Days
Vehicle control
JBI-2174, 3 mg/kg QD
JBI-2174, 10 mg/kg, QD
JBI-2174, 30 mg/kg, BID
Isotype control
Atezolizumab, 200 ug

MC38/hPD-L1 intracranial tumors

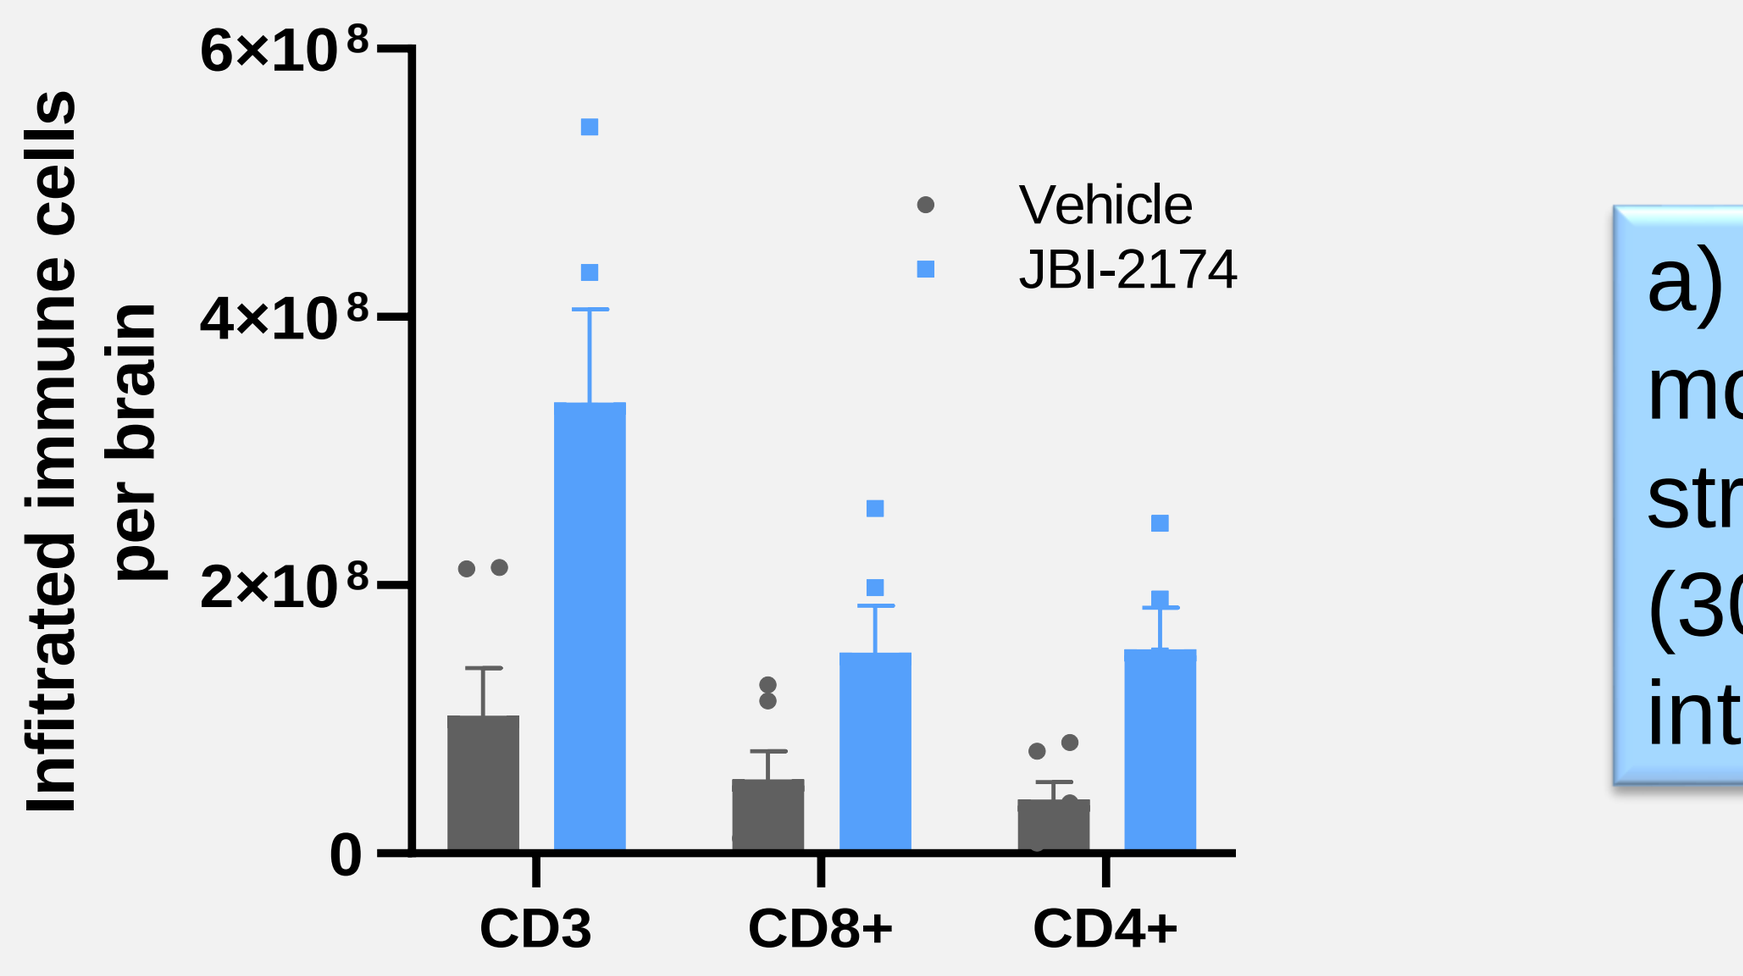
Infitrated immune cells per brain
Vehicle
JBI-2174
CD3
CD8+
CD4+

a) JBI-2174 is highly efficacious in MC38/hPD-L1 orthotopic model and enhances survival by 35% b) comparable or stronger efficacy to Atezolizumab was observed in syngeneic (30 mg/kg, BID, PO) model ; c) leads strong infiltration of T-cells into the brain tumor

JBI-2174 is well tolerated in NHP toxicology studies and shows brain exposure

a **Non-human primate toxicology**

- No morbidity or mortality
- No change in body weight
- No test article related changes in hematology or serum chemistry
- No gross or histopathological changes in any organ
- Well tolerated beyond efficacious exposure

b

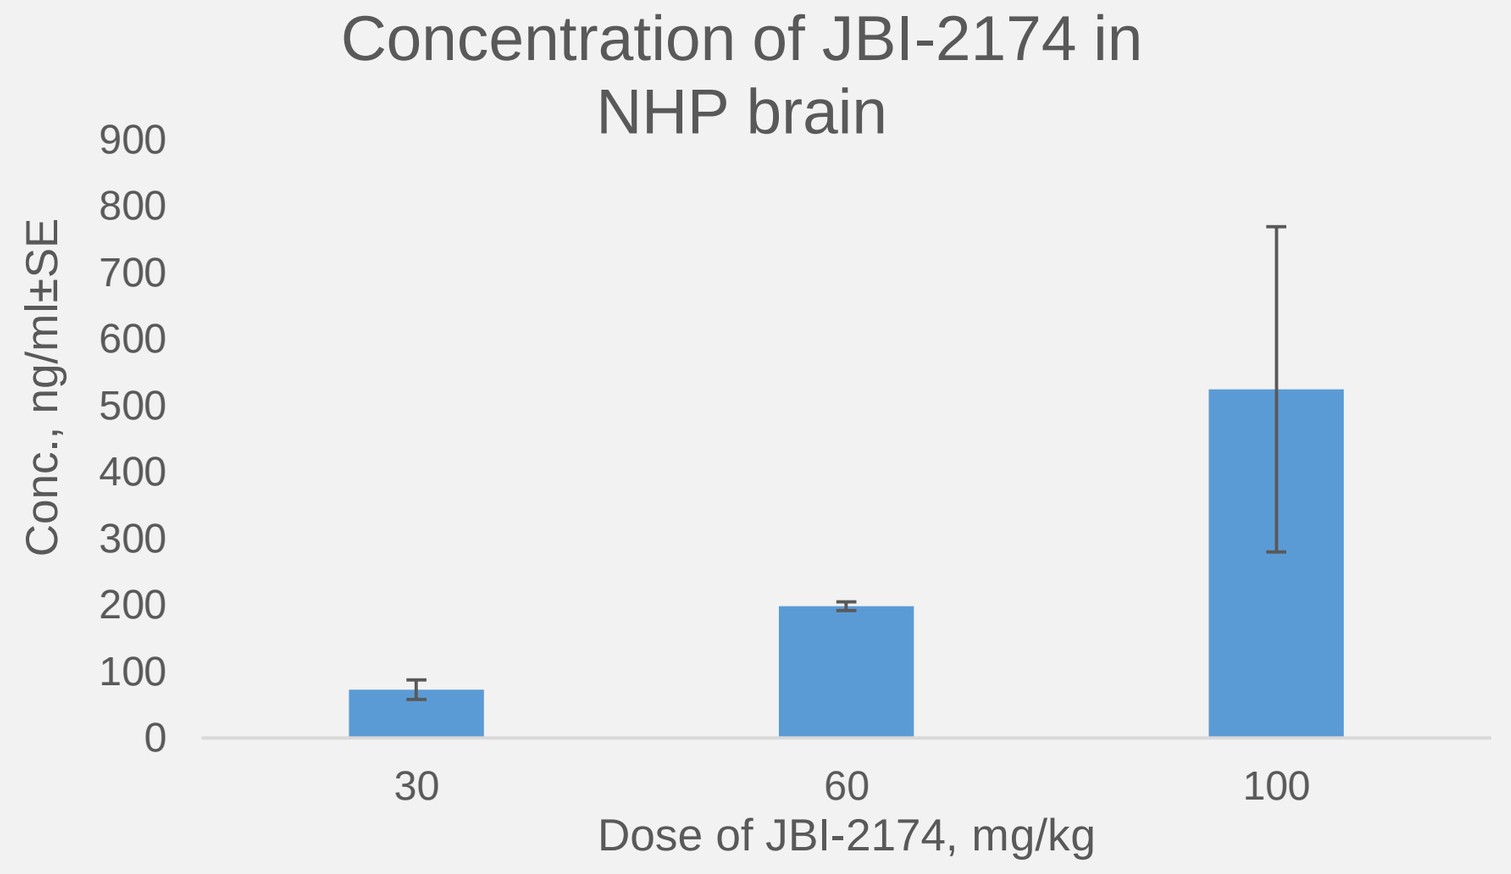
Concentration of JBI-2174 in NHP brain
Conc., ng/ml±SE
30
60
100
Dose of JBI-2174, mg/kg

Non-GLP toxicology studies clearly indicate that JBI-2174 is well tolerated and it crosses BBB

Conclusions

- JBI-2174 is a novel, potent and selective PD-L1 inhibitor that shows strong and sustained brain exposure by oral administration. Structural and other mechanistic studies have confirmed that JBI-2174 binds and works similar to anti-PD-L1 antibodies. In vivo studies have demonstrated that JBI-2174 shows strong tumor growth inhibition and enhances tumor infiltrating T-lymphocytes.
- Exploratory toxicology studies in NHP clearly demonstrate that JBI-2174 is well tolerated and it effectively penetrated blood brain barrier
- IND-enabling studies are being initiated with JBI-2174

Contact: luca.rastelli@jubilanttx.com

Do Not Post