Novel, brain penetrant small molecule inhibitor of PD-L1 for targeting glioblastoma and brain metastasis

Dhanalakshmi Sivanandhan[1], Sridharan Rajagopal[1], Chandru Gajendran[1], Naveen Sadhu M[1], Zainuddin Mohd[1], Ramachandraiah Gosu[1], **Luca Rastelli[2]**

[1]Jubilant Therapeutics India Limited, Bangalore, India. [2]Jubilant Therapeutics Bedminster, NJ.

Jubilant Therapeutics, Bedminster, NJ

Poster # 1376

Abstract # 1376

SUMMARY

Background: The PD-1/PD-L1 molecular pathway is one of the primary mechanisms of immune evasion deployed by cancer cells and anti-PD-L1 antibodies (mAbs) restore T-cell proliferation and enhanced tumor cell killing which has been shown to result in clinically revolutionary efficacy in many tumor types. Unfortunately, this is not the case in CNS tumors where these mAbs have failed to show improved responses and survival. It appears that one of the main reasons for this is the poor brain penetrance of these mAbs and clinical evidence suggests that increasing brain penetrance results in better efficacy. Therefore, small molecule inhibitors of PD-L1 with enhanced tumor and brain penetrance could become highly valuable in CNS cancer therapy. In addition, small molecules with optimized oral bioavailability and short half-life, can address other known limitations of the PD-1/PD-L1 antibodies, including the well-known immune-breakthrough toxicities. JBI-2174 is an orally available selective small molecule inhibitor with similar binding and mechanism of action as anti-PD-L1 antibodies. JBI-2174 is highly brain penetrant and shows comparable efficacy to approved mAbs in preclinical studies.

Materials and methods: Structure based drug design was used to design PD-L1 inhibitors; potency of these inhibitors was assessed in an *in-vitro* TR-FRET assay. Reporter assays and ex-vivo co-culture assays were used to assess T-cell proliferation and function. Pharmacokinetics were performed in multiple pre-clinical species to derive at bioavailability and brain penetration. In vivo efficacy was assessed in mice syngeneic and orthotopic models.

Results: Our lead molecule JBI-2174 showed strong *in vitro* IC_{50} of 1.5 nM in TR-FRET assay that measures interaction between PD-1 and PD-L1 and also induced dimerization of PD-L1 in cell based assay. This molecule also augmented T-cell response as measured by IFN-γ activation in a cancer cell-PBMC based co-culture assay. Competition study between anti-PD-L1 blocking antibody and x-ray co-crystallisation studies clearly demonstrate that JBI-2174 has similar finger-print on PD-L1 as the anti-PD-L1 antibodies. More importantly, JBI-2174 showed good oral bioavailability across pre-clinical species and strong and sustained brain exposure (0.66 to 2.1 fold plasma vs. brain ratio). JBI-2174 showed comparable efficacy to the anti-PD-L1 antibody Atezolizumab in hPD-L1/MC38 syngeneic and orthotopic models by oral administration. Toxicological studies conducted in non-human primates clearly show that the molecule is well tolerated at exposures much higher than efficacious exposure and that it crosses BBB.

Conclusion: Oral administration and brain exposure of JBI-2174 provides an attractive option to be used in the treatment of glioblastoma and other solid tumors with brain metastasis. IND-enabling studies are being initiated for this compound.

JBI-2174 is highly potent against PD-L1

a

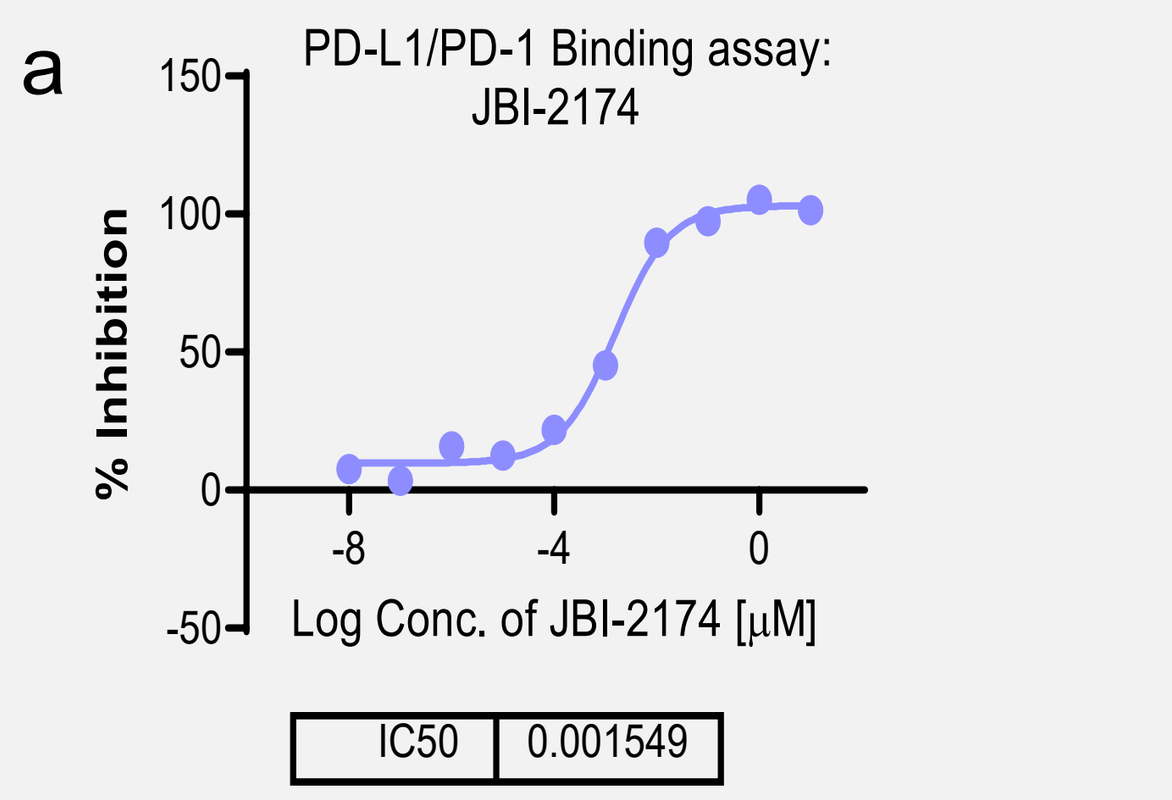
PD-L1/PD-1 Binding assay:
JBI-2174
% Inhibition
Log Conc. of JBI-2174 [µM]
IC50 0.001549

b

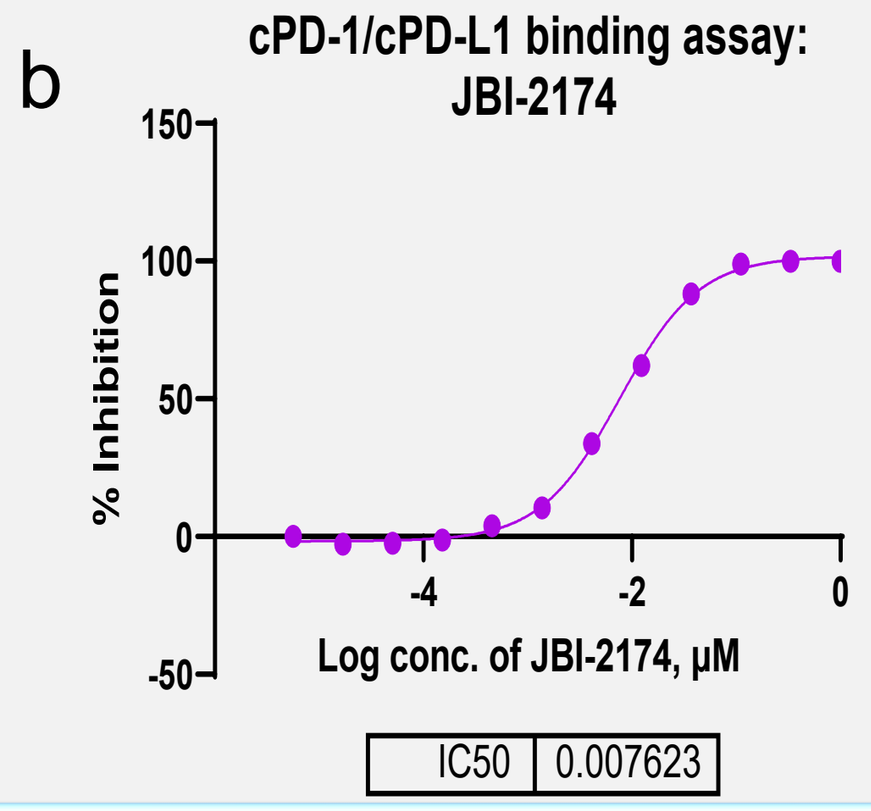
cPD-1/cPD-L1 binding assay:
JBI-2174
% Inhibition
Log conc. of JBI-2174, µM
IC50 0.007623

In vitro potency of JBI-2174 against (a) human PD-1/PD-L1 was assessed by HTRF and (b) cyno PD-1/PD-L1 was assessed by ELISA based Binding Activity (BPS Biosciences)

JBI-2174 binds to PD-L1 leading to dimerization of PD-L1

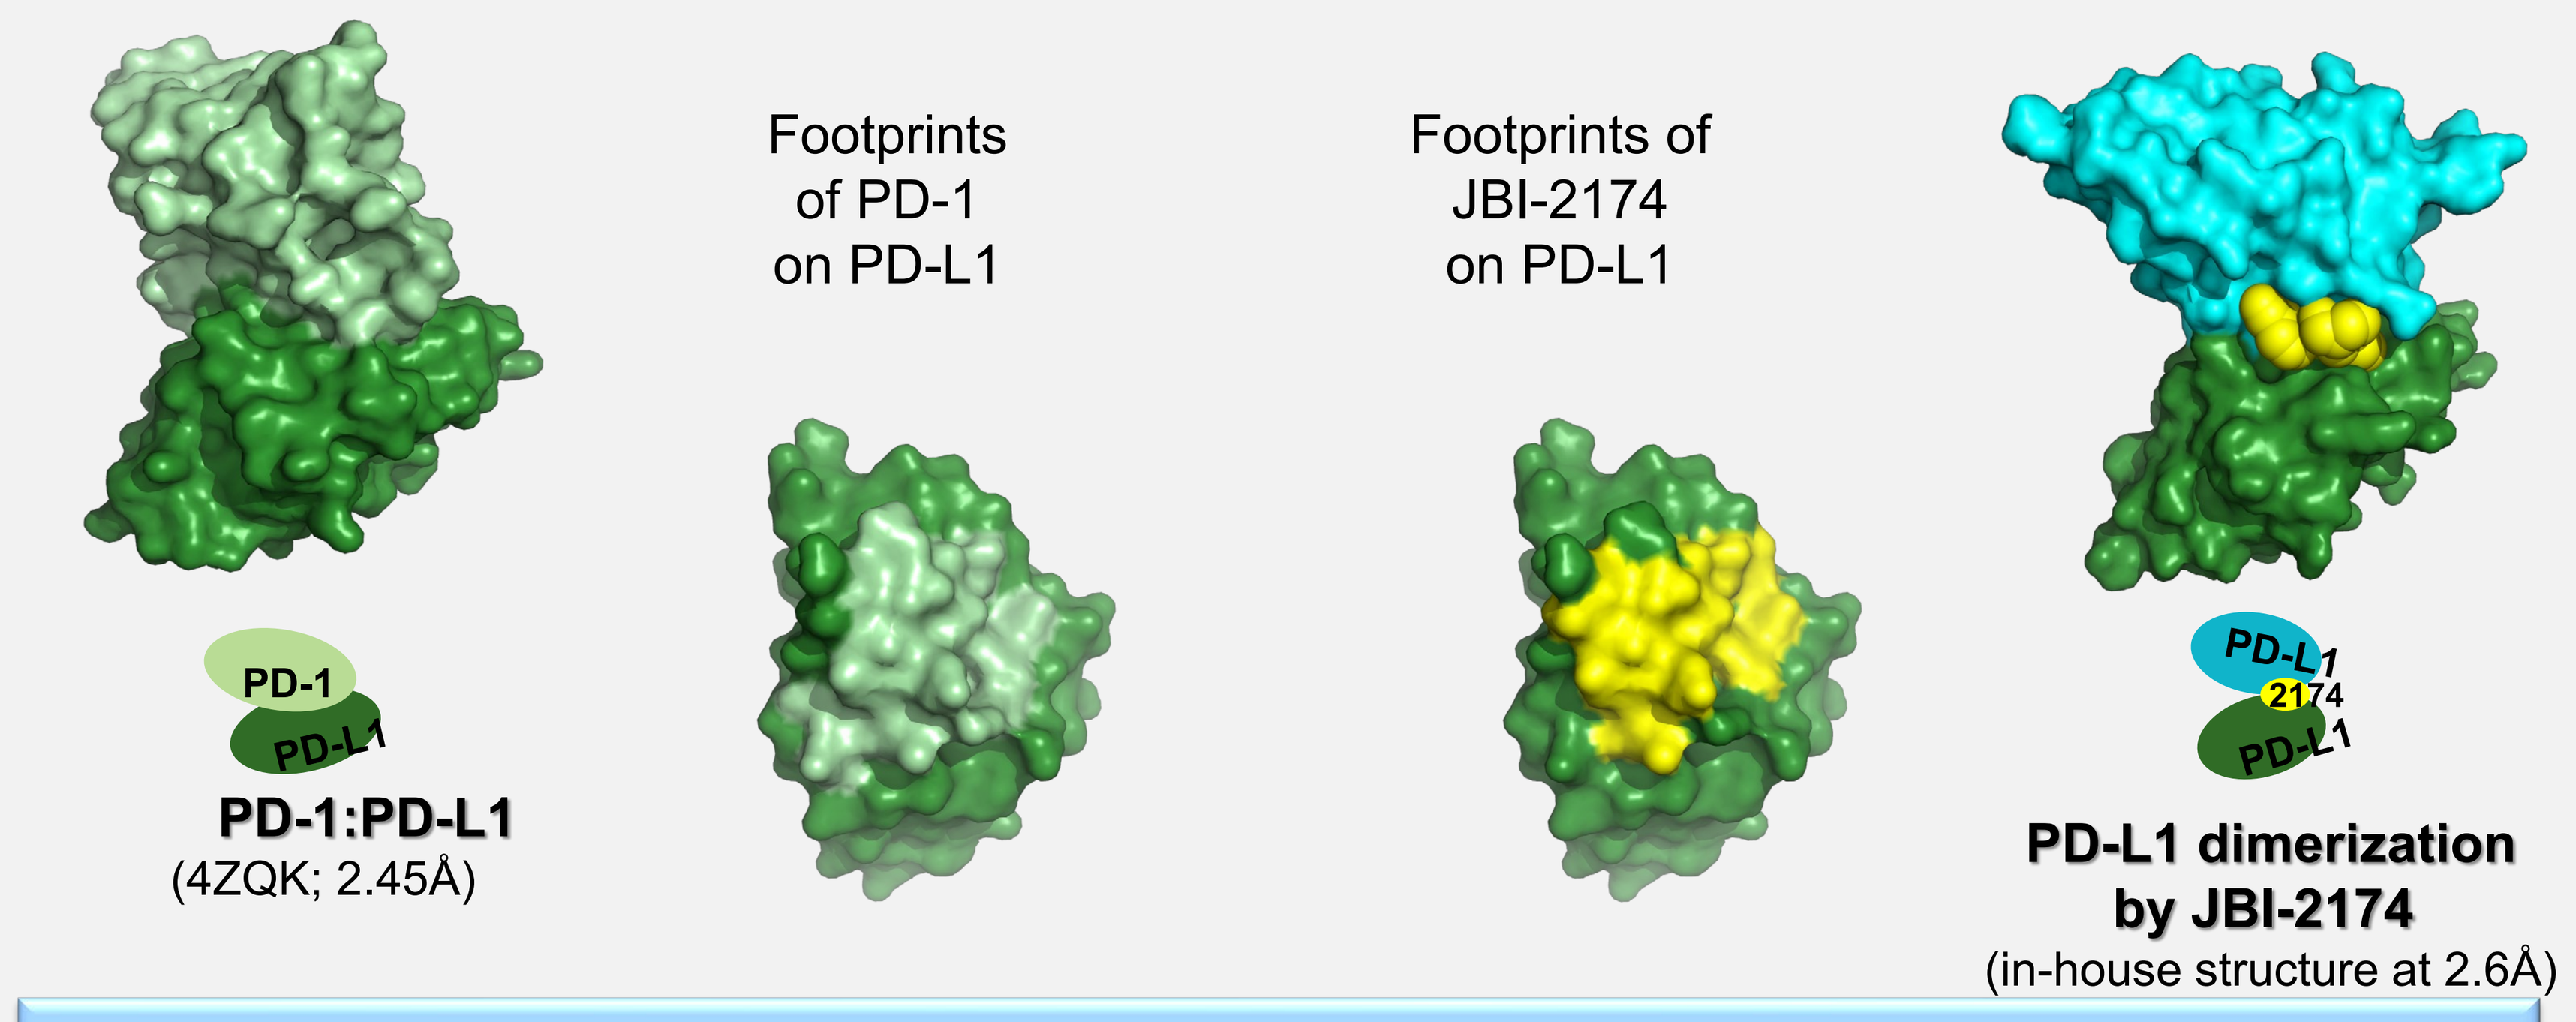
Footprints of PD-1 on PD-L1
Footprints of JBI-2174 on PD-L1
PD-1
PD-L1
2174
PD-1:PD-L1
(4ZQK; 2.45Å)
PD-L1 dimerization by JBI-2174
(in-house structure at 2.6Å)

- JBI-2174 binds selectively to PD-L1 and leads to dimerization of PD-L1; JBI-2174 covers 65% of PD-1 footprint, similar to mAbs

JBI-2174 interrupts PD-1/PD-L1 signaling

a

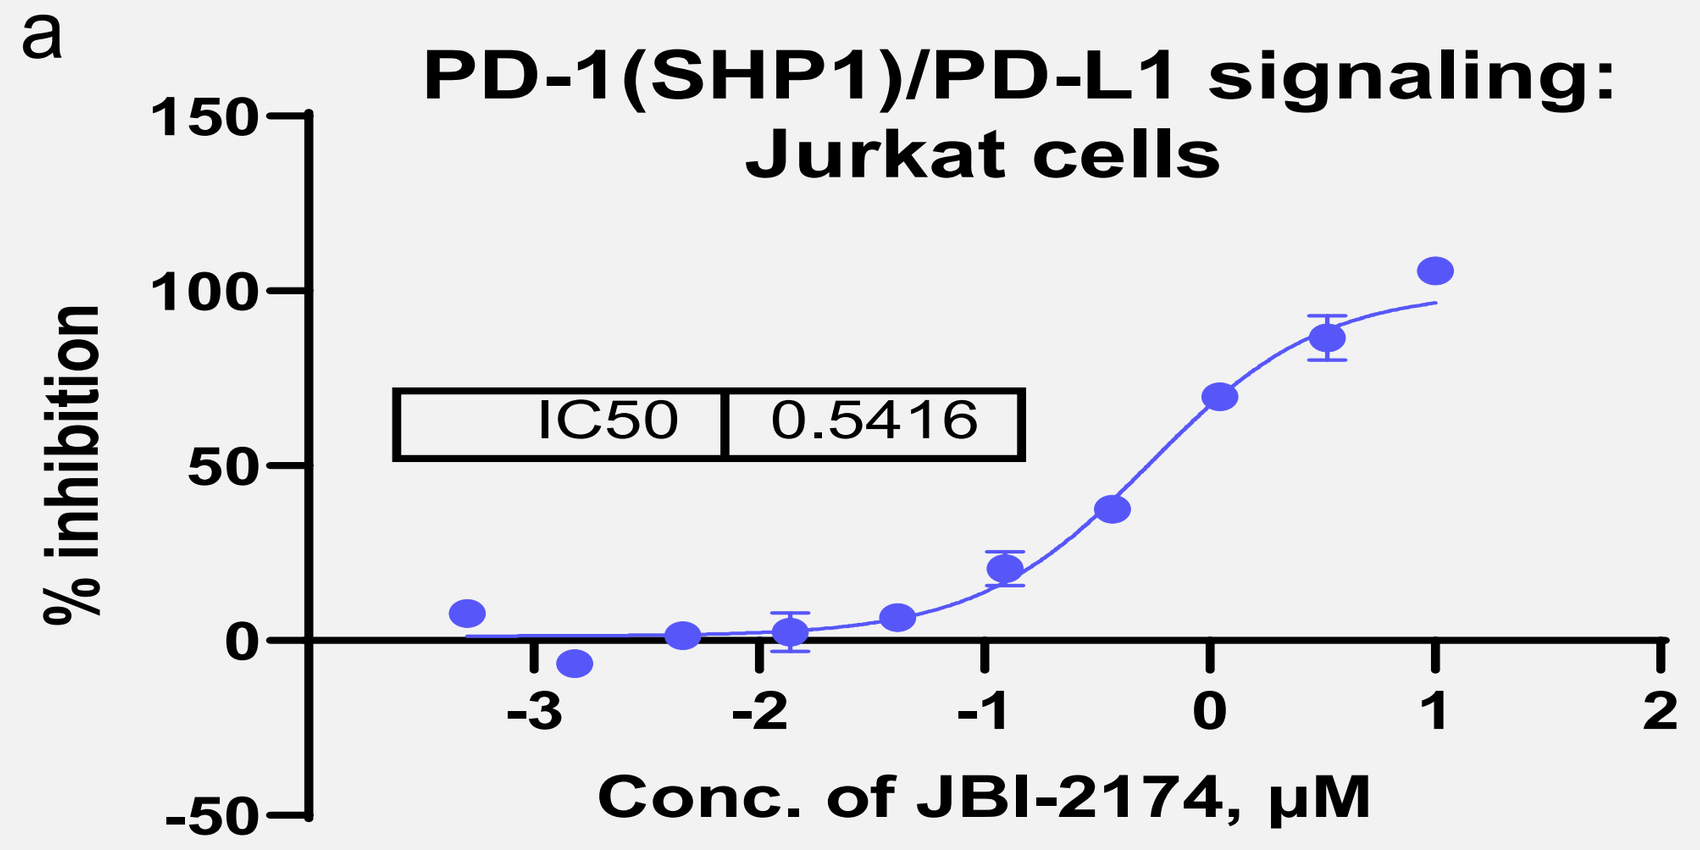
PD-1(SHP1)/PD-L1 signaling:
Jurkat cells
% inhibition
IC50 0.5416
Conc. of JBI-2174, µM

b

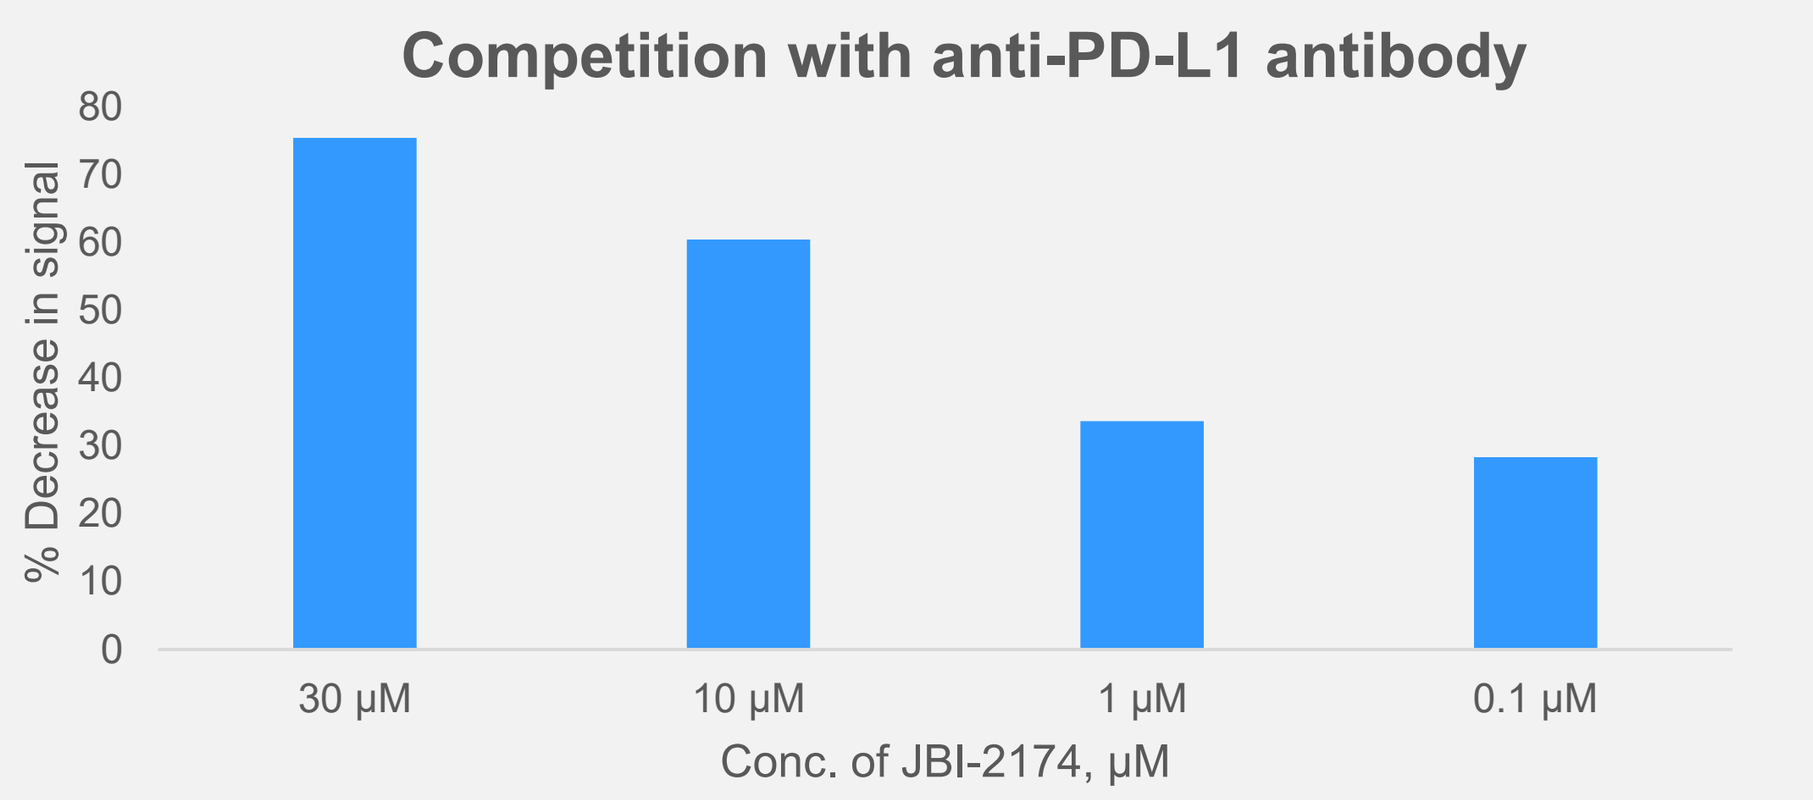
Competition with anti-PD-L1 antibody
% Decrease in signal
30 µM
10 µM
1 µM
0.1 µM
Conc. of JBI-2174, µM

Note: a) Jurkat cells expressing the PD-1 and SHP-1 proteins are fused to a enzyme fragment complementation (EFC) PathHunter PD-L1/PD-1 signaling assay; JBI-2174 incubation time: 2h followed by incubation with PD-1 (SHP-1) cells. b) JBI-2174 incubation time: 1.5 h

a)JBI-2174 interrupts PD-L1 binding with PD-1 b) Hs746T treated with JBI-2174 and incubated with Anti-PDL-1 PE (blocking) antibody for 30 min. Mean fluorescence intensity of PE was analyzed by FACS

JBI-2174 is efficacious in MC38/hPD-L1 syngeneic model

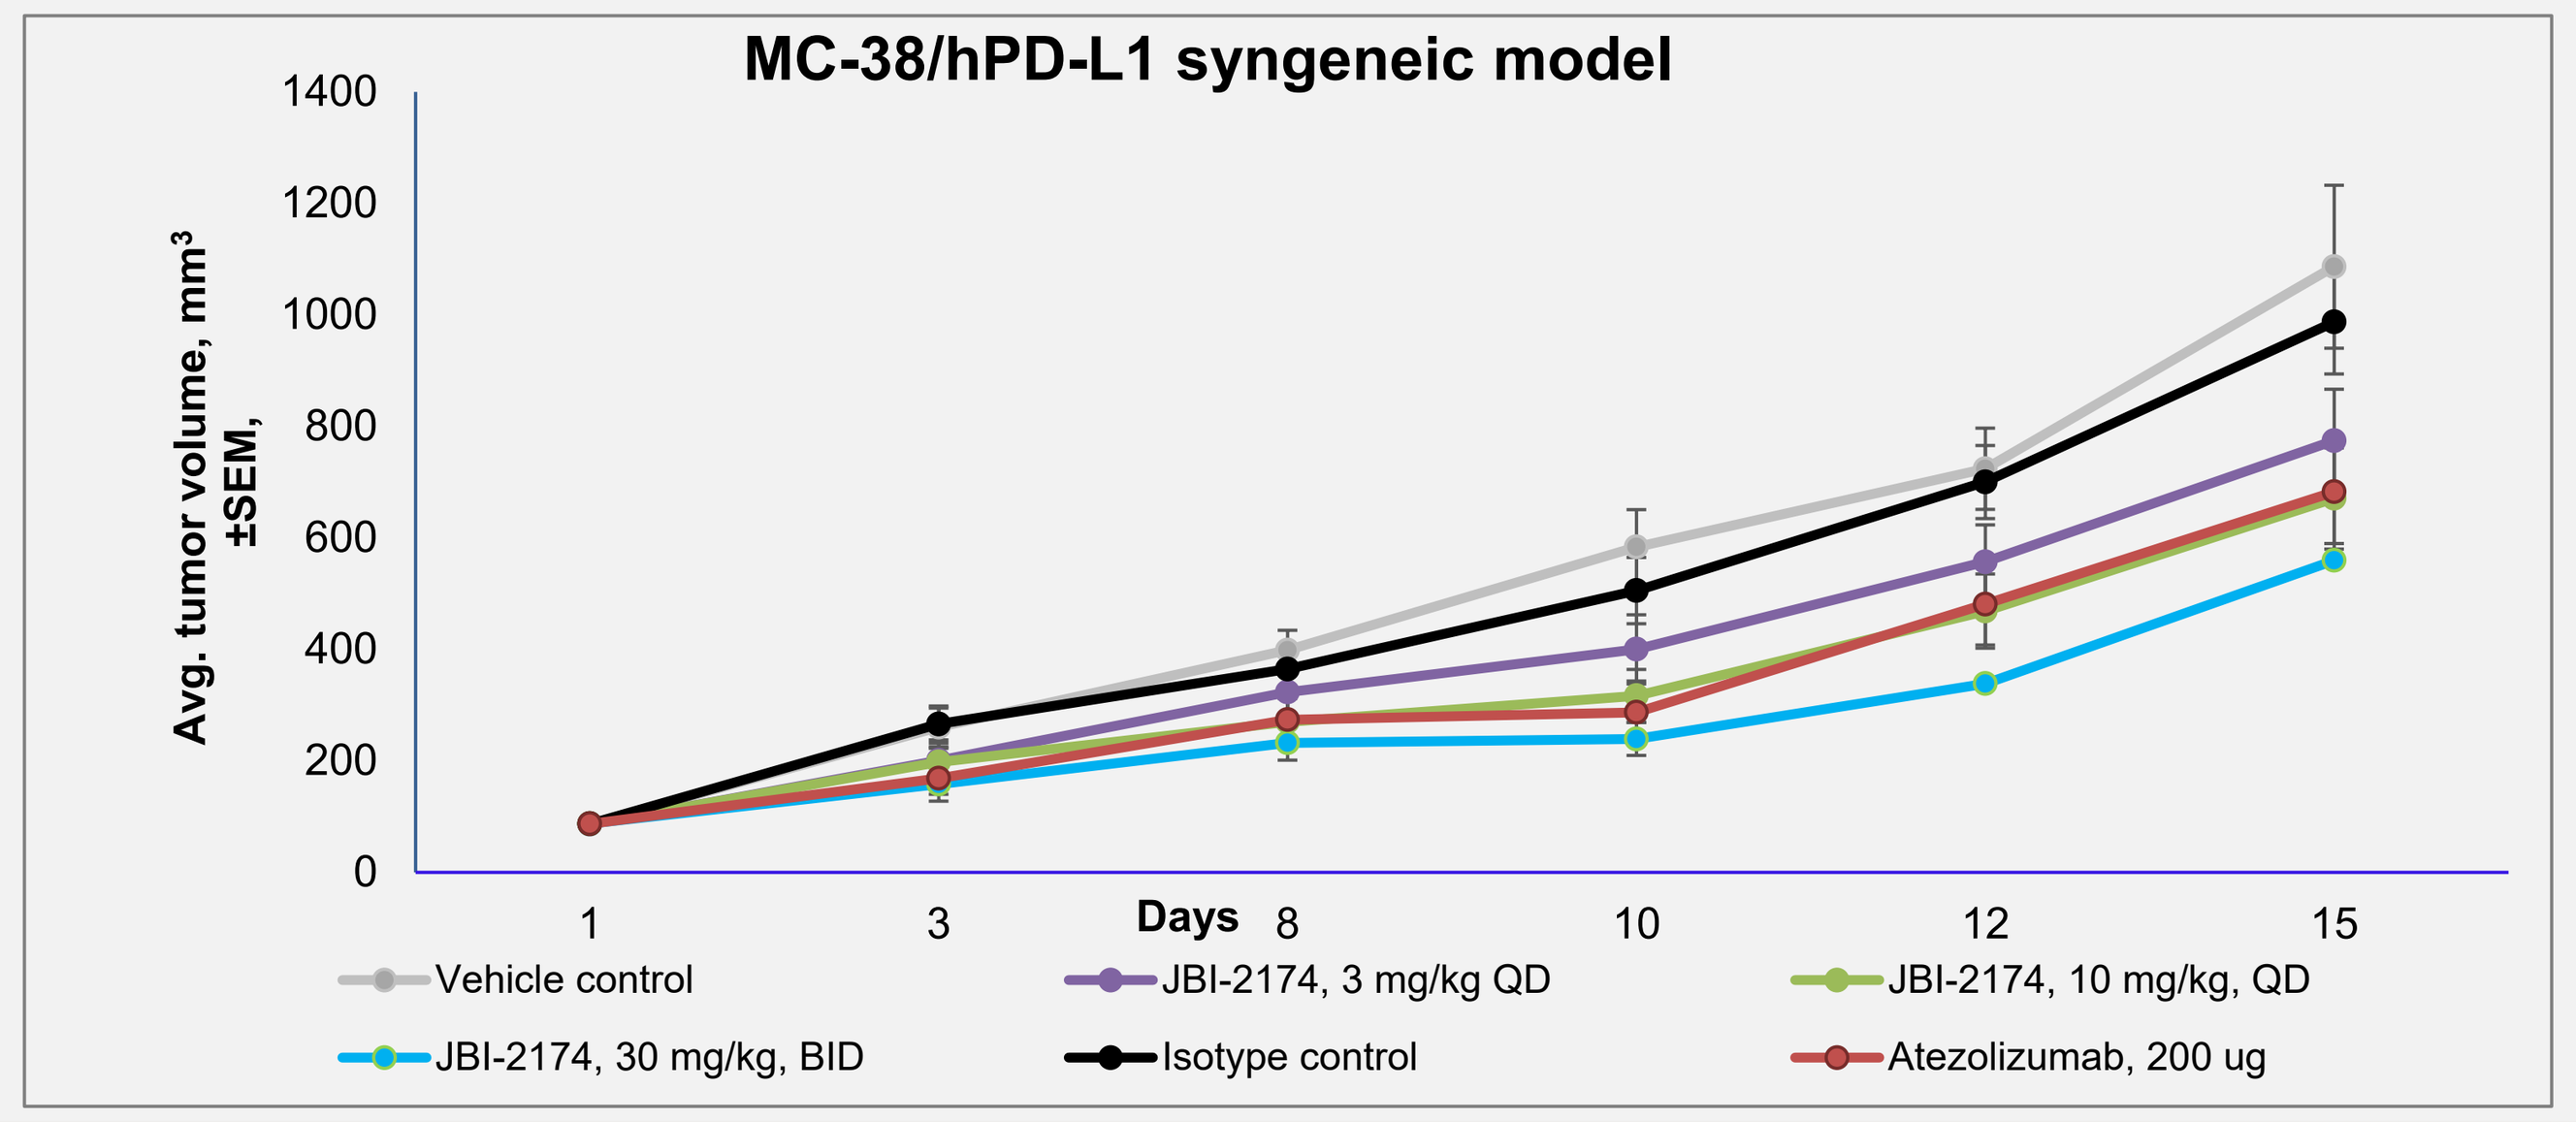
MC-38/hPD-L1 syngeneic model
Avg. tumor volume, mm³ ±SEM,
Days
Vehicle control
JBI-2174, 3 mg/kg QD
JBI-2174, 10 mg/kg, QD
JBI-2174, 30 mg/kg, BID
Isotype control
Atezolizumab, 200 ug

JBI-2174 shows stronger or comparable efficacy to Atezolizumab in syngeneic (30 mg/kg, BID, PO) model

JBI-2174 is highly brain-penetrant and enhances TILs in MC38/hPD-L1 orthotopic model

a

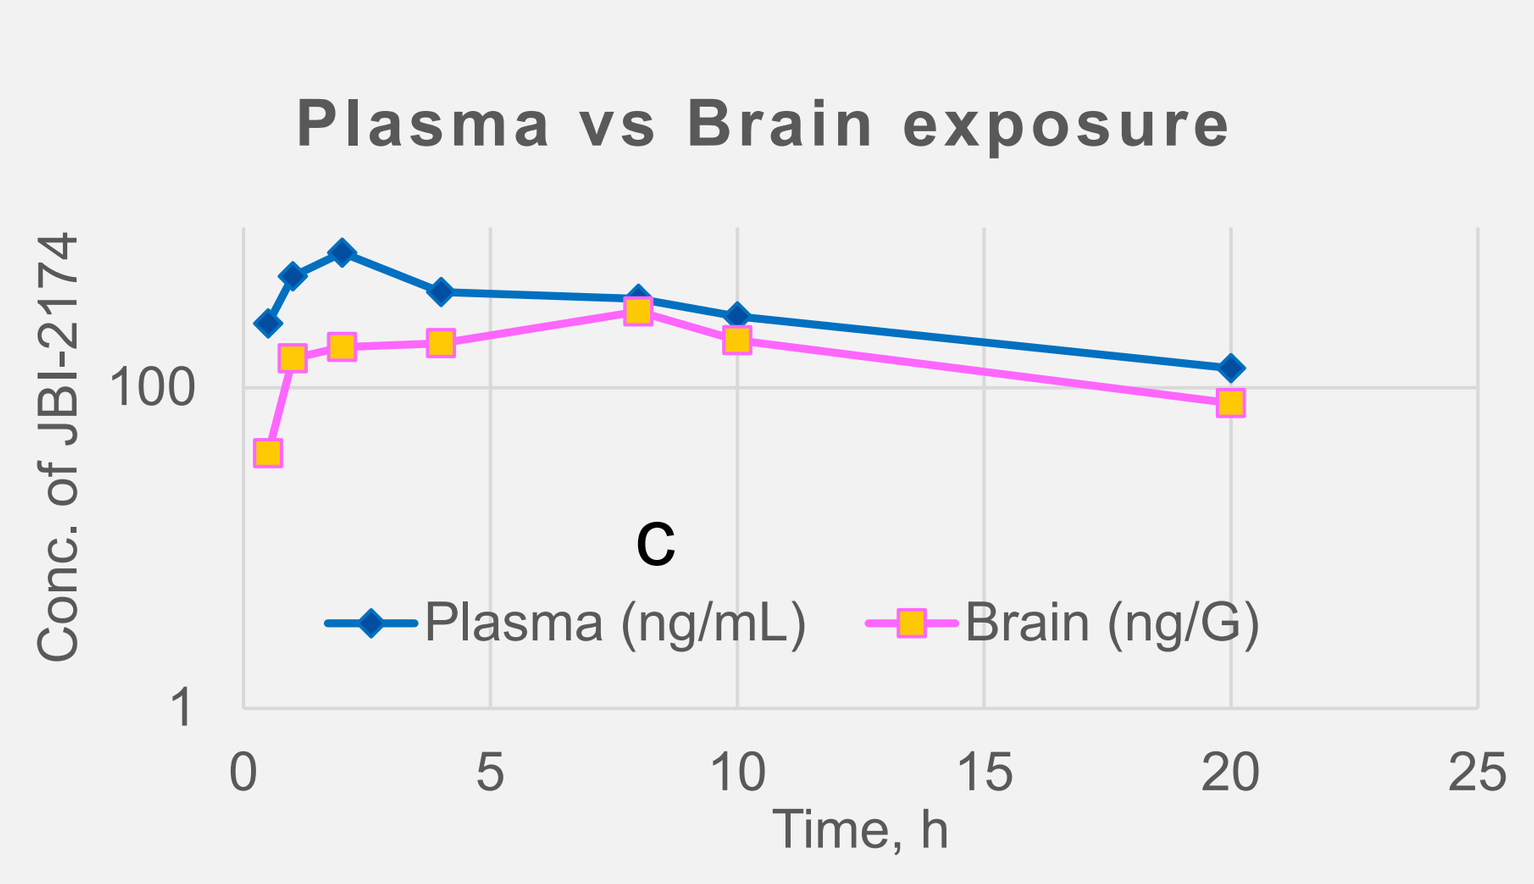
Exposure in SD rat
Plasma vs Brain exposure
Conc. of JBI-2174
c
Plasma (ng/mL)
Brain (ng/G)
Time, h

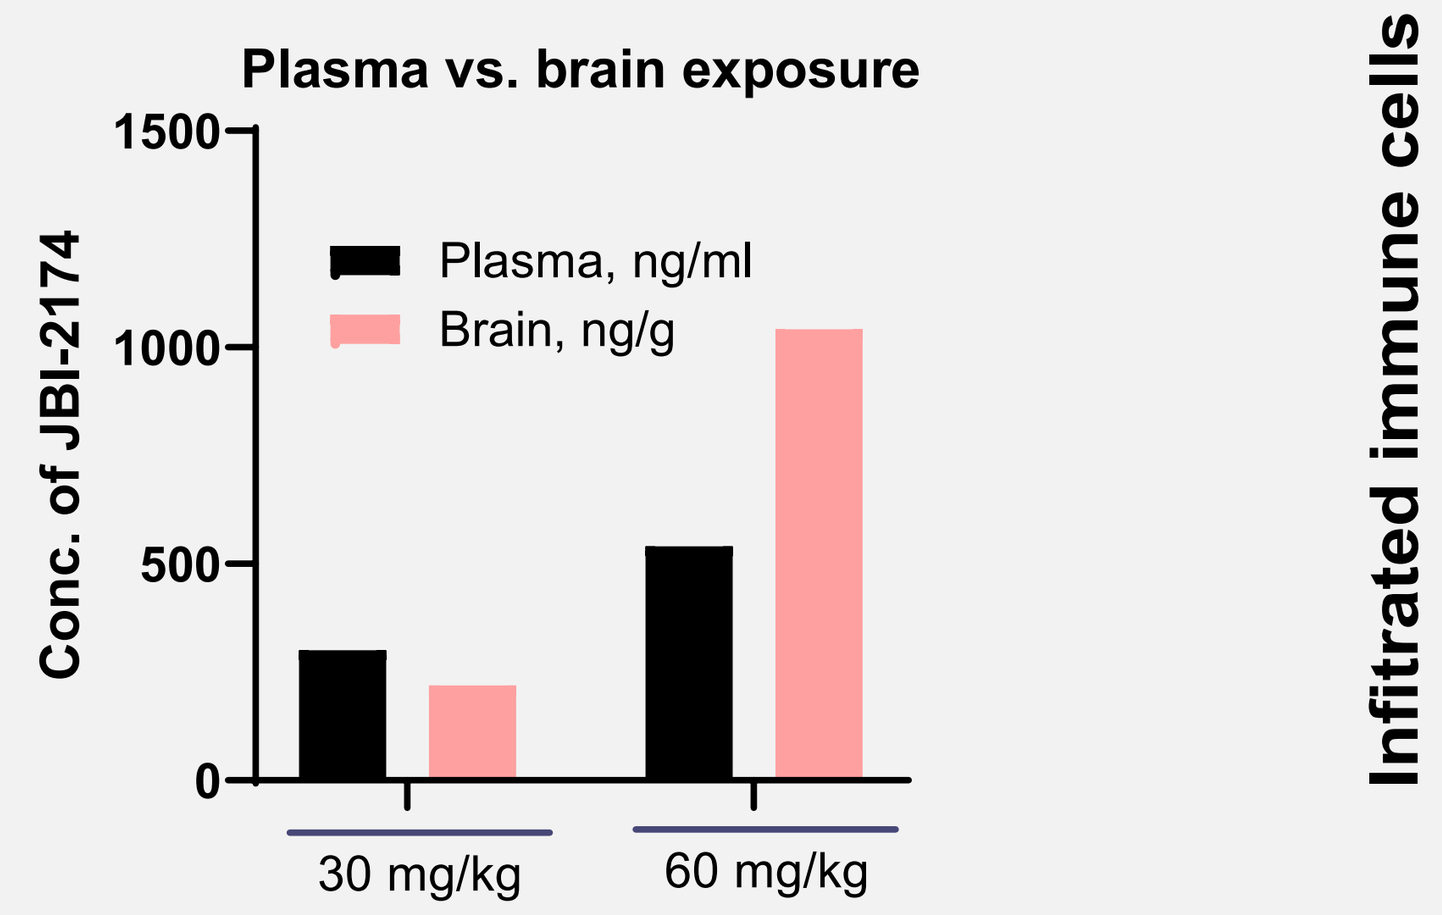
Exposure in mice
Plasma vs. brain exposure
Conc. of JBI-2174
Plasma, ng/ml
Brain, ng/g
30 mg/kg
60 mg/kg

Note: Conc. given are 24 h post dose

b

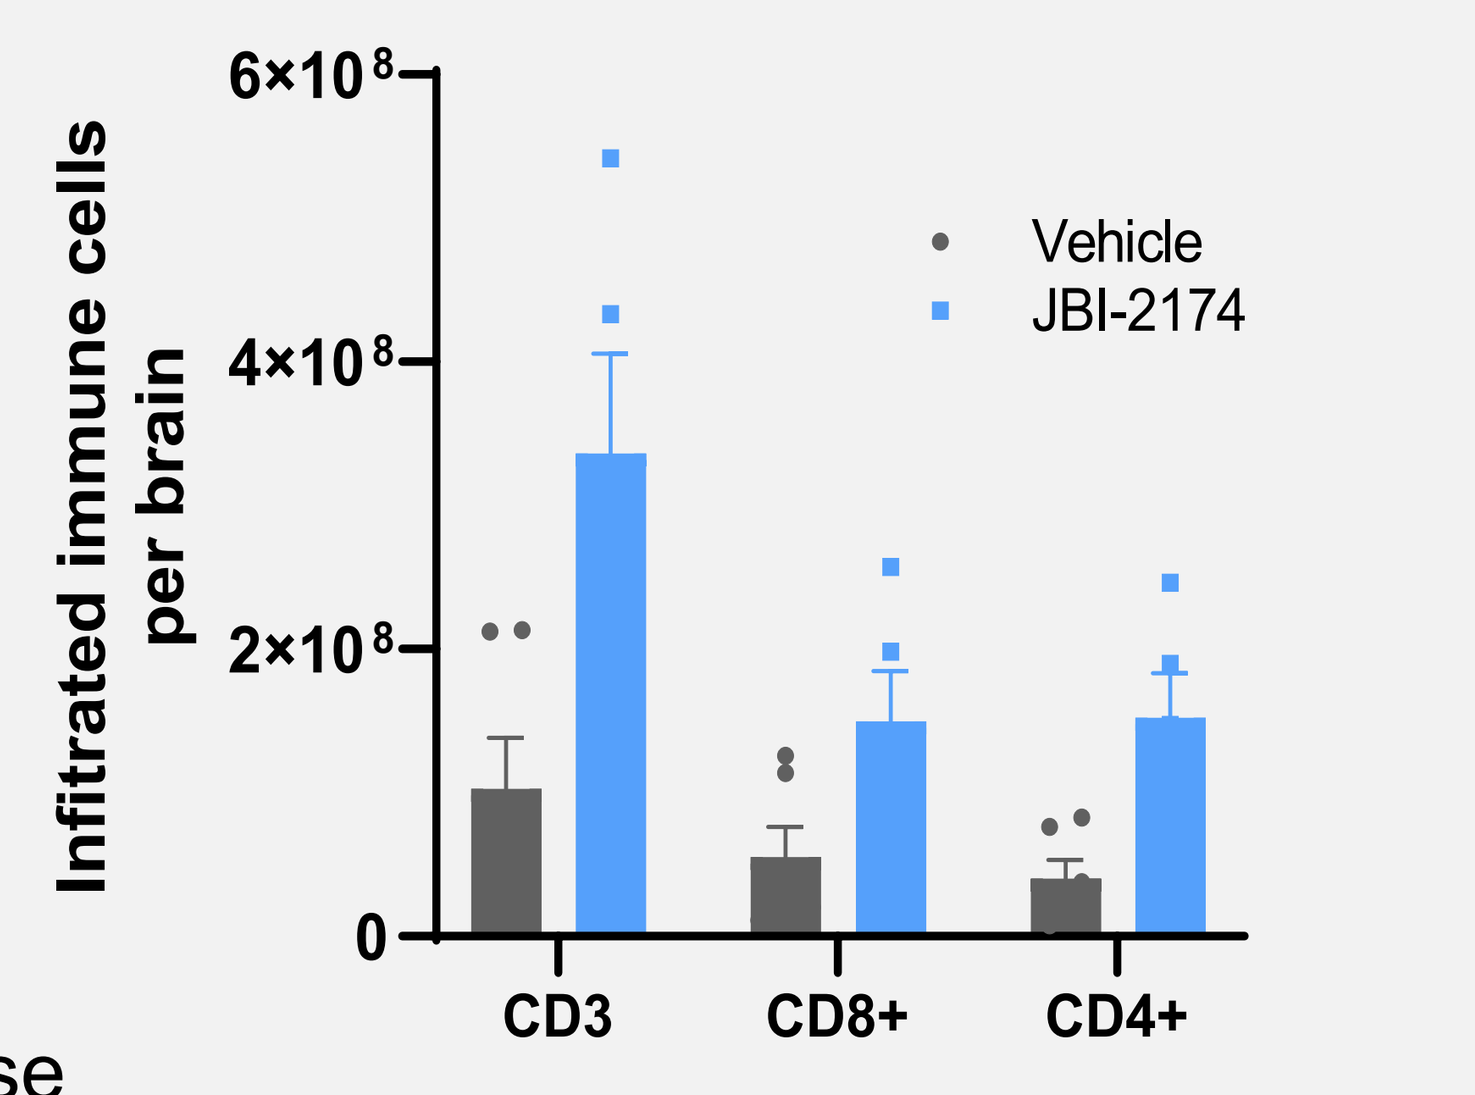
MC38/hPD-L1 intracranial tumors
Infitrated immune cells per brain
Vehicle
JBI-2174
CD3
CD8+
CD4+

a) JBI-2174 shows strong and sustained brain exposure in Rat and Mice and b) enhances tumor infiltrating lymphocytes in intracranial tumors

JBI-2174 is well tolerated in NHP toxicology studies and crosses BBB

a **14-day repeat dose non-human primate toxicology**

- No morbidity or mortality
- No change in body weight
- No test article related changes in hematology or serum chemistry
- No gross or histopathological changes in any organ

b

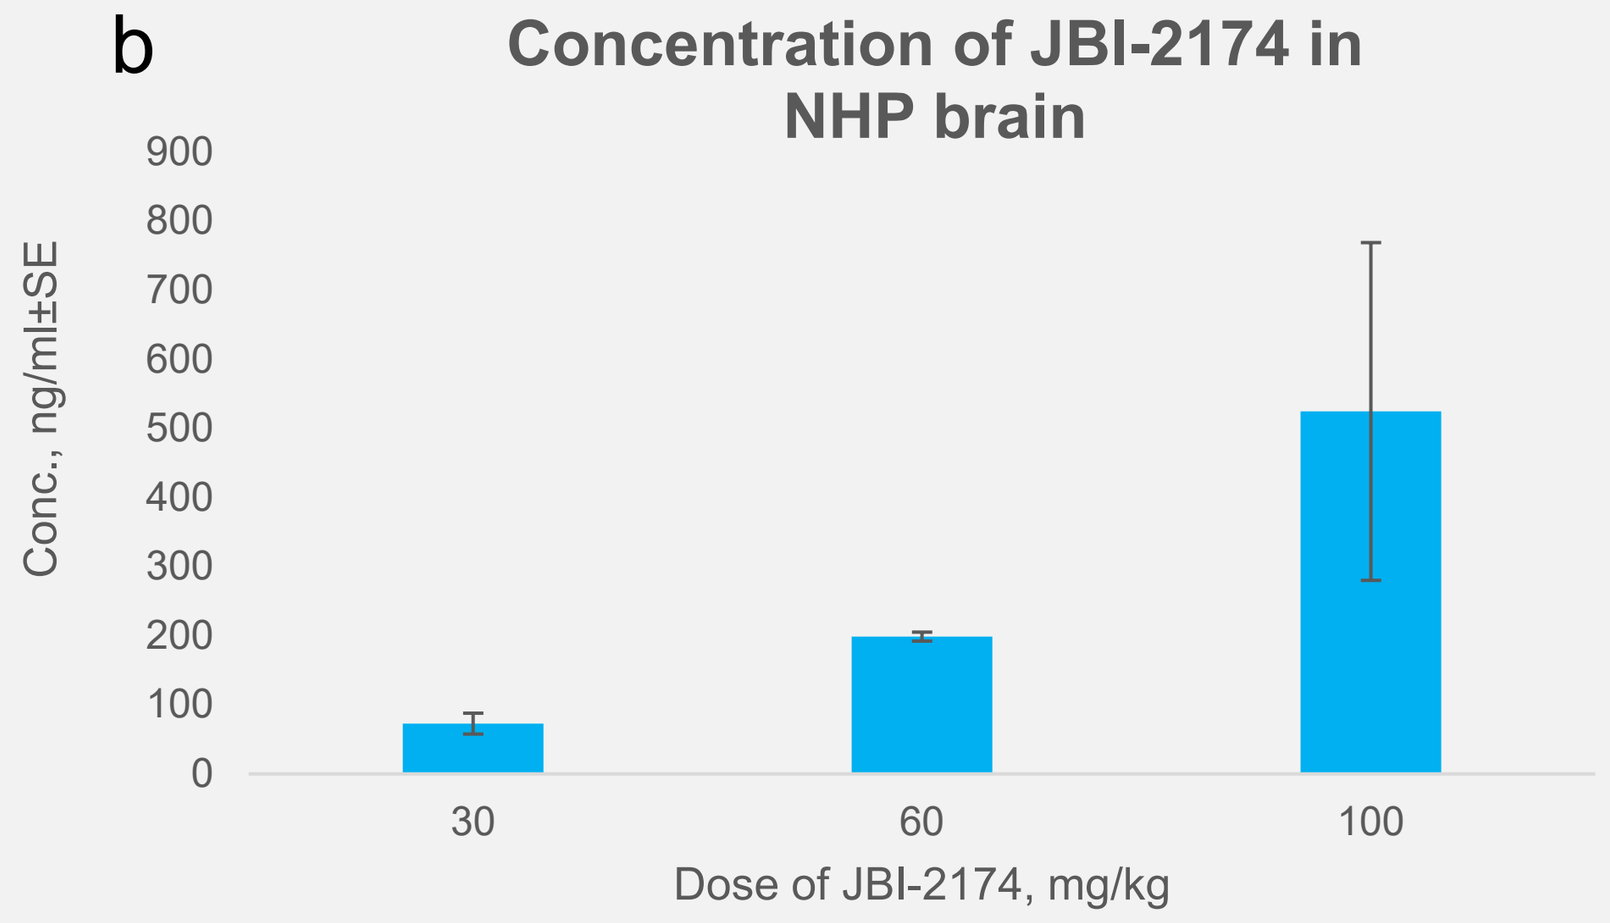
Concentration of JBI-2174 in NHP brain
Conc., ng/ml±SE
Dose of JBI-2174, mg/kg

Non-GLP toxicology studies clearly indicate that JBI-2174 is well tolerated and it crosses BBB

Conclusions

- JBI-2174 is a novel, potent and selective PD-L1 inhibitor that shows strong and sustained brain exposure by oral administration. Structural and other mechanistic studies have confirmed that JBI-2174 binds and works similar to anti-PD-L1 antibodies. In vivo studies have demonstrated that JBI-2174 shows strong tumor growth inhibition and enhances tumor infiltrating T-lymphocytes.
- Exploratory toxicology studies in NHP clearly demonstrate that JBI-2174 is well tolerated and it effectively penetrated blood brain barrier
- IND-enabling studies are being initiated with JBI-2174

Acknowledgements

We would like to thank Amir Siddiqui M, Annasaheb S.kalange, Santosh Vishwakarma, Ashwini Narayan, Pratima Deshpande (Jubilant Biosys Ltd, Bangalore, India) for assistance in conducting the experiments.

Contact: luca.rastelli@jubilanttx.com / Dhanalakshmi.Sivanandhan@jubilanttx.com

Do Not Post