Novel, gut-restricted small molecule inhibitor of PD-L1 for targeting GI cancers

Dhanalakshmi Sivanandhan[1], Sridharan Rajagopal[1], Chandru Gajendran[1], Naveen Sadhu M[1], Zainuddin Mohd[1], Ramachandraiah Gosu[1], **Luca Rastelli[2]**

[1]Jubilant Therapeutics India Limited, Bangalore, India. [2]Jubilant Therapeutics Bedminster, NJ.

Jubilant Therapeutics, Bedminster, NJ

Poster # 1386

Abstract # 1386

SUMMARY

Background: The PD-1/PD-L1 molecular pathway is one of the primary mechanisms of immune evasion deployed by cancer cells and several anti-PD-L1 antibodies (mAbs) have been approved for the treatment of multiple cancers, including cancers of the GI tract and liver. In such cancers, one of the issues observed is broad-spectrum autoimmune toxicities, including pneumonitis, and others. primarily owing to their long systemic half-life. Therefore, small molecule inhibitors of PD-L1 with gut restricted exposure should potentially be able overcome these toxicities. JBI-1527 is a small molecule PD-L1 inhibitor with similar binding and mechanism of action as anti-PD-L1 antibodies that shows much higher exposure in gastrointestinal tract and liver as compared to plasma. It shows comparable efficacy as approved mAbs in preclinical studies.

Materials and methods: Structure based drug design was used to design PD-L1 inhibitors; potency of these inhibitors was assessed in an *in-vitro* TR-FRET assay. Reporter assays and ex-vivo co-culture assays were used to assess T-cell proliferation and function. Pharmacokinetics studies were performed in multiple pre-clinical species to assess tissue distribution. In vivo efficacy was assessed in partially humanized mice efficacy models.

Results: JBI-1527 showed strong *in vitro* IC_{50} of 2.8 nM in TR-FRET assay that measures interaction between PD-1 and PD-L1 and led to stabilization of PD-L1 protein as measured by thermal shift assay. This molecule also augmented T-cell co-inhibitory signalling as observed by Jurkat cell/SHP-1 assay. Competition study between anti-PD-L1 blocking antibody suggested that JBI-1527 finger-printing on PD-L1 is similar to mAbs. X-ray crystal structure studies clearly demonstrated that JBI-1527 caused dimerization of PD-L1. More importantly, JBI-1527 showed favourable gastrointestine localised pharmacokinetic profile with high exposure in colon, jejunum, duodenum, ileum (11 to >280 fold vs. plasma) as compared to plasma in preclinical species when dosed orally. JBI-1527 showed comparable efficacy to the anti-PD-L1 antibody Atezolizumab in hPD-L1/MC38 syngeneic and orthotopic models by oral administration and is well tolerated at efficacious doses.

Conclusion: Gastrointestine localised pharmacokinetic profile of JBI-1527 provides an attractive option to be used in the treatment of colon cancer, HCC and other GI-related cancers with minimal systemic toxicity as compared to mAbs

Biochemical characterization

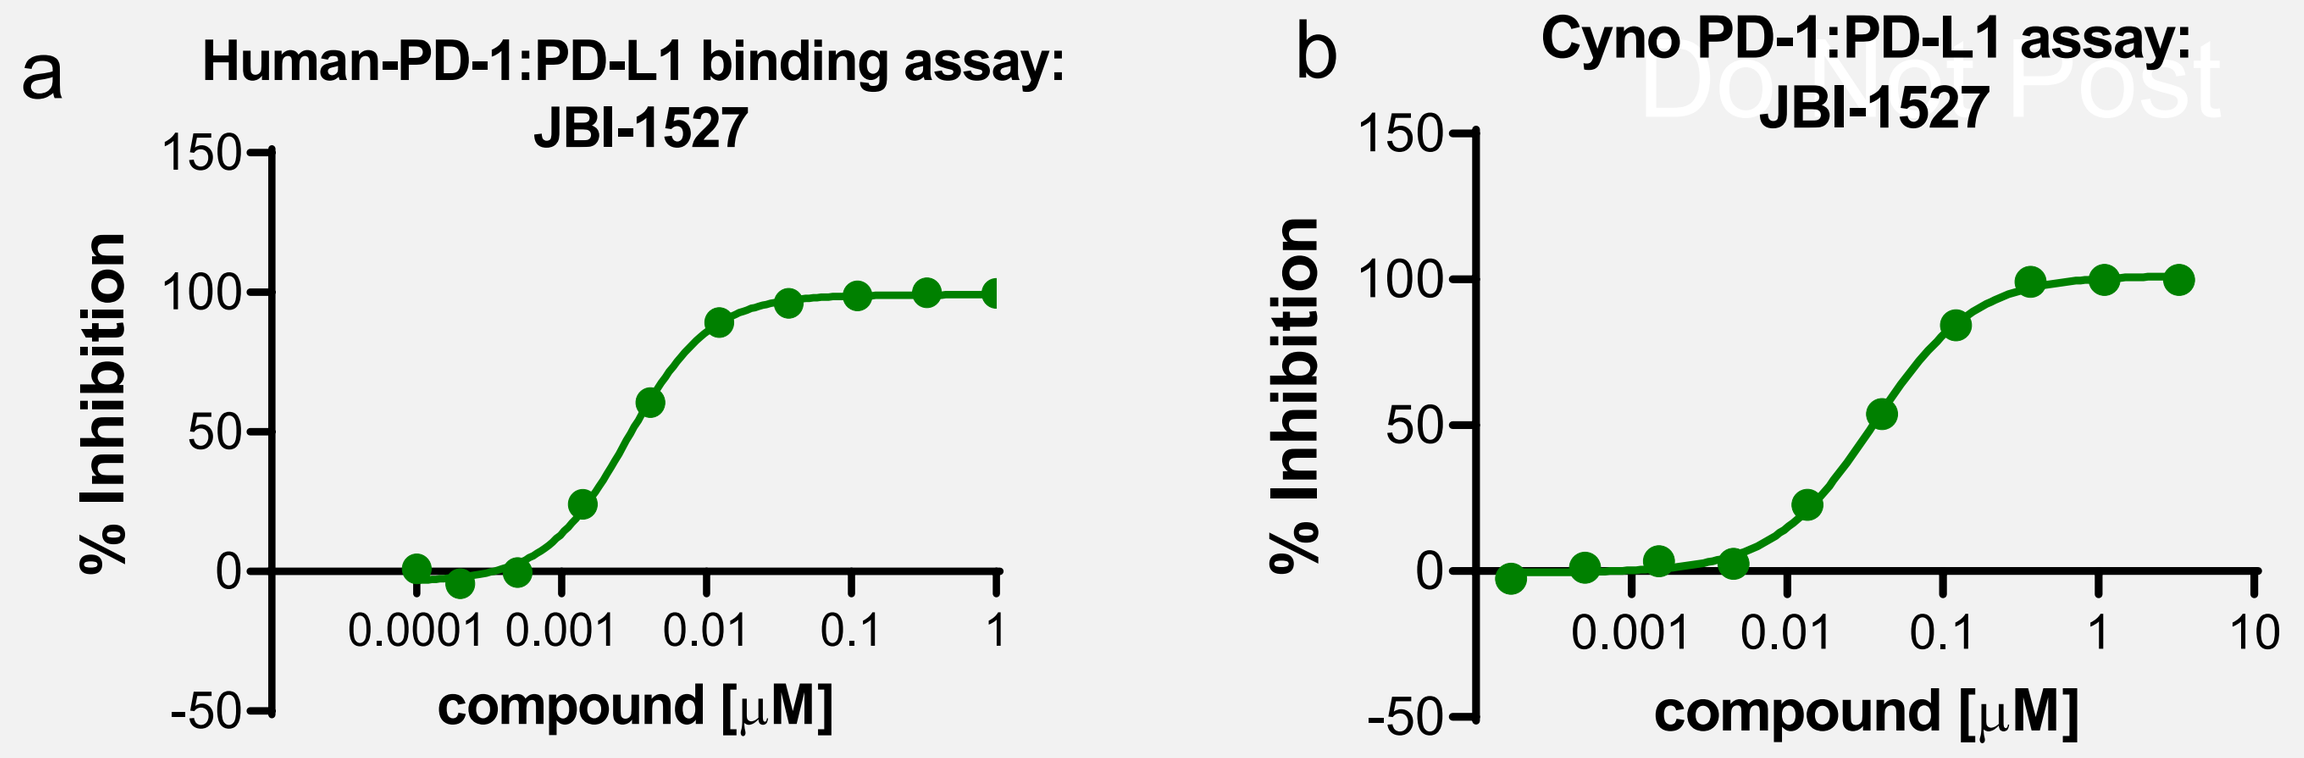
a
Human-PD-1:PD-L1 binding assay: JBI-1527
% Inhibition
compound [μM]
b
Cyno PD-1:PD-L1 assay: JBI-1527
% Inhibition
compound [μM]

	JBI-1527
IC50	0.002874

	JBI-1527
IC50	0.03597

In vitro potency of JBI-1527 against (a) human PD-1/PD-L1 assessed by HTRF and (b) cyno PD-1/PD-L1 was assessed by ELISA based Binding Activity assay done at BPS Biosciences

JBI-1527 competes with anti-PD-L1 blocking antibody

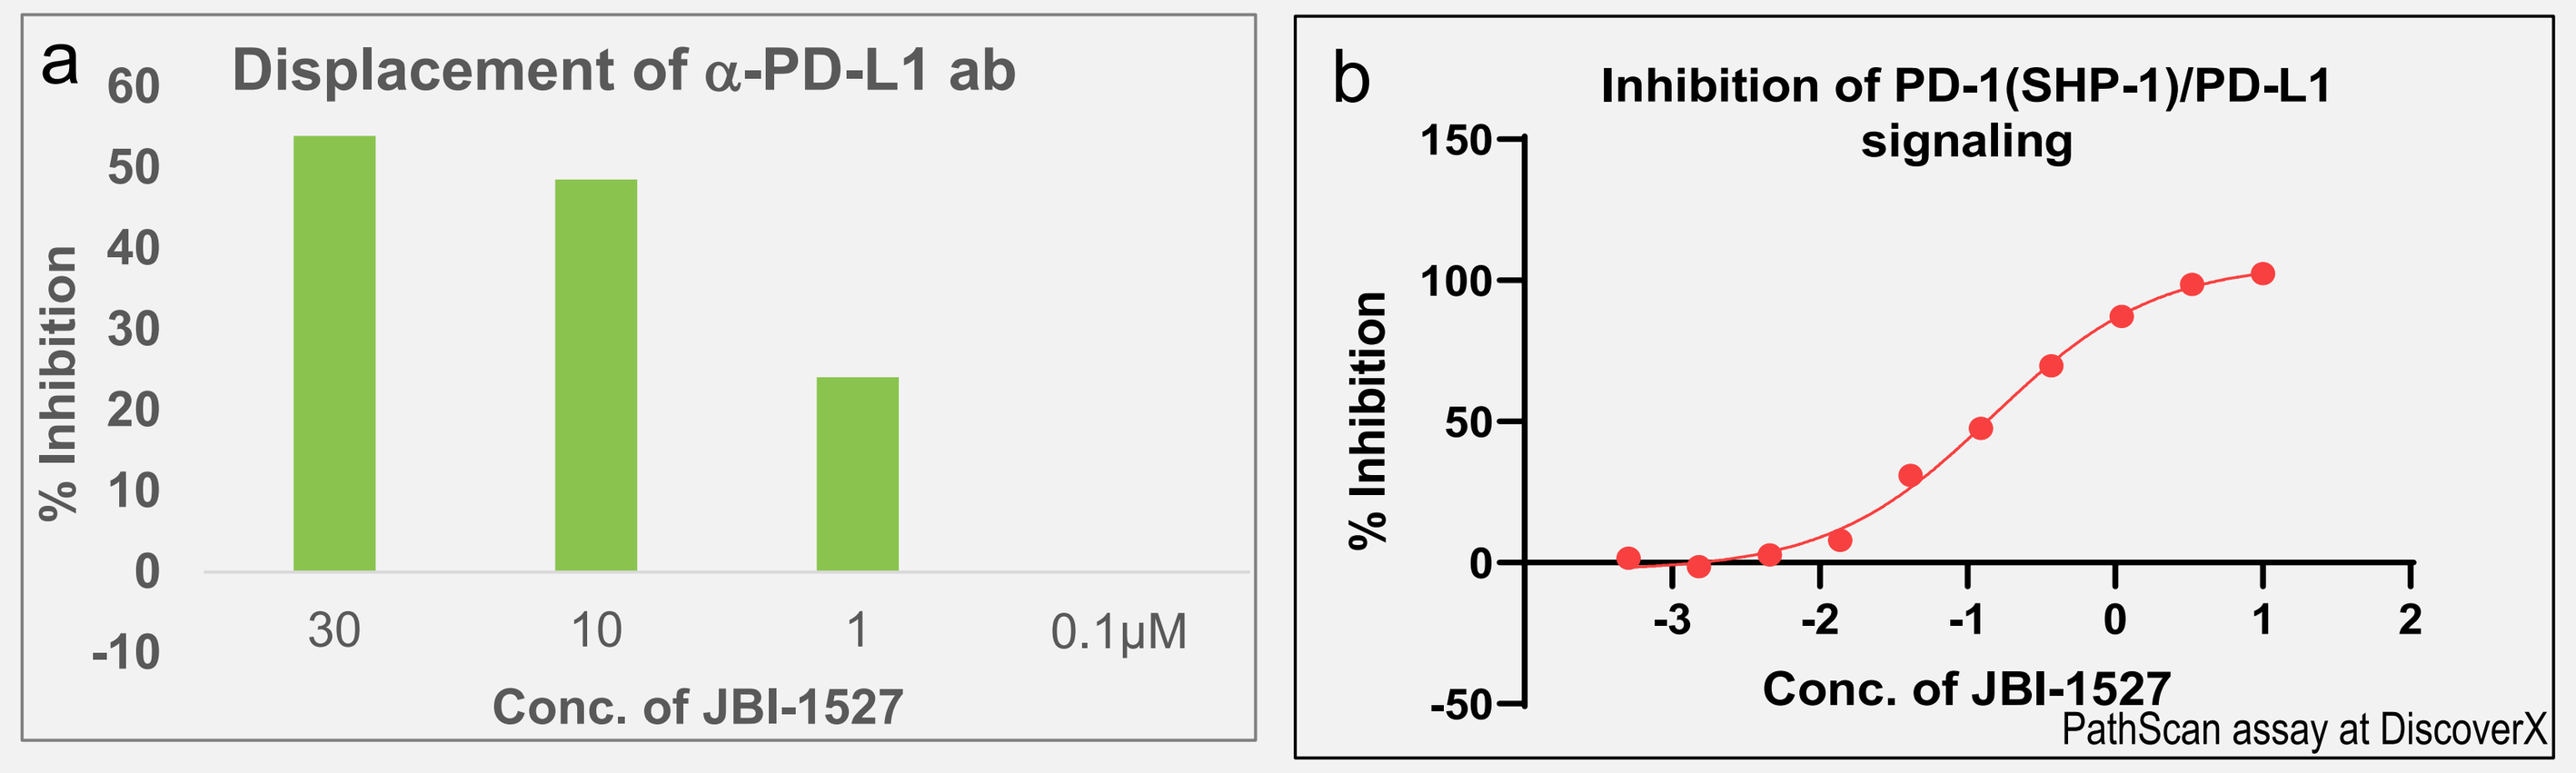
a
Displacement of α-PD-L1 ab
% Inhibition
30
10
1
0.1μM
Conc. of JBI-1527
b
Inhibition of PD-1(SHP-1)/PD-L1 signaling
% Inhibition
Conc. of JBI-1527
PathScan assay at DiscoverX

Note: a) Jurkat cells expressing the PD-1 and SHP-1 proteins are fused to a enzyme fragment complementation (EFC) PathHunter PD-L1/PD-1 signaling assay; JBI-1527 incubation time: 2h followed by incubation with PD-1 (SHP-1) cells. B) JBI-1527 incubation time: 1.5 h

a) Hs746T treated with JBI-1527 and incubated with Anti-PDL-1 PE (blocking) antibody for 30 min. Mean fluorescence intensity of PE was analyzed by FACS; b) JBI-1527 interrupts PD-L1 binding with PD-1

Modulation of BioMAP panel

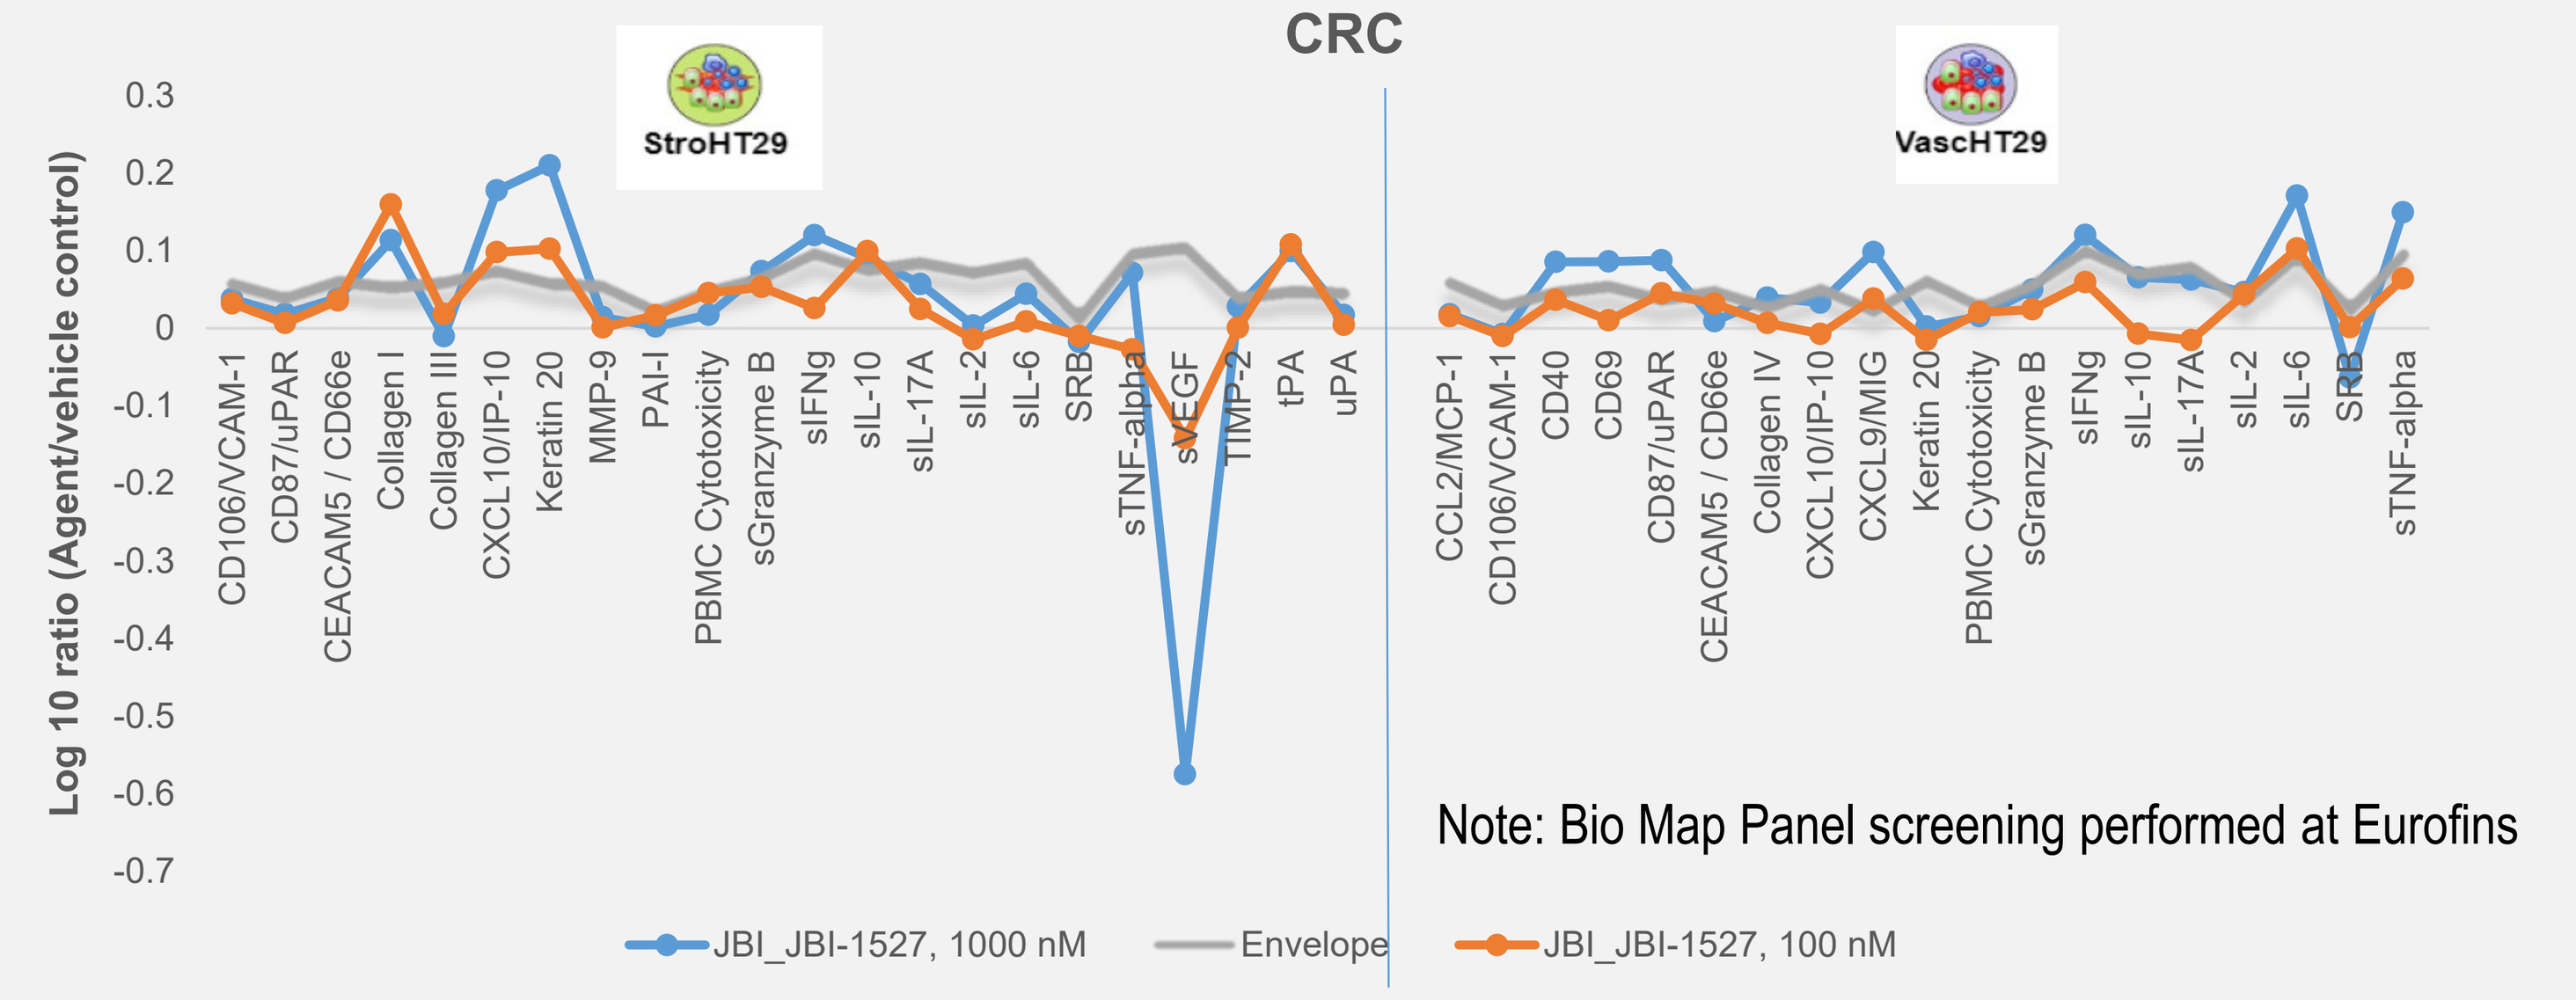
CRC
StroHT29
VascHT29
Log 10 ratio (Agent/vehicle control)
JBI_JBI-1527, 1000 nM
Envelope
JBI_JBI-1527, 100 nM

Note: Bio Map Panel screening performed at Eurofins

System	Cell Types
StroHT29	HT-29 colorectal adenocarcinoma cell line + Peripheral blood mononuclear cells + Primary human fibroblasts
VascHT29	HT-29 colorectal adenocarcinoma cell line + Peripheral blood mononuclear cells + Primary human endothelial cells

Biological and Disease relevance	JBI-1527	Pembrolizumab
Inflammation related	sTNF-a, IP10 ↑	sTNF-a
Matrix remodeling	Col-1↑	-
Angiogenesis	sVEGF↓, tPA↑	sVEGF↓
Tumor related	Keratin 20↑	-
Immune related	sIFNg, sIL6 ↑	sIFNg, sIL6

Modulation of BioMAP panel by JBI-1527 is comparable to Pembrolizumab

JBI-1527 leads to dimerization of PD-L1

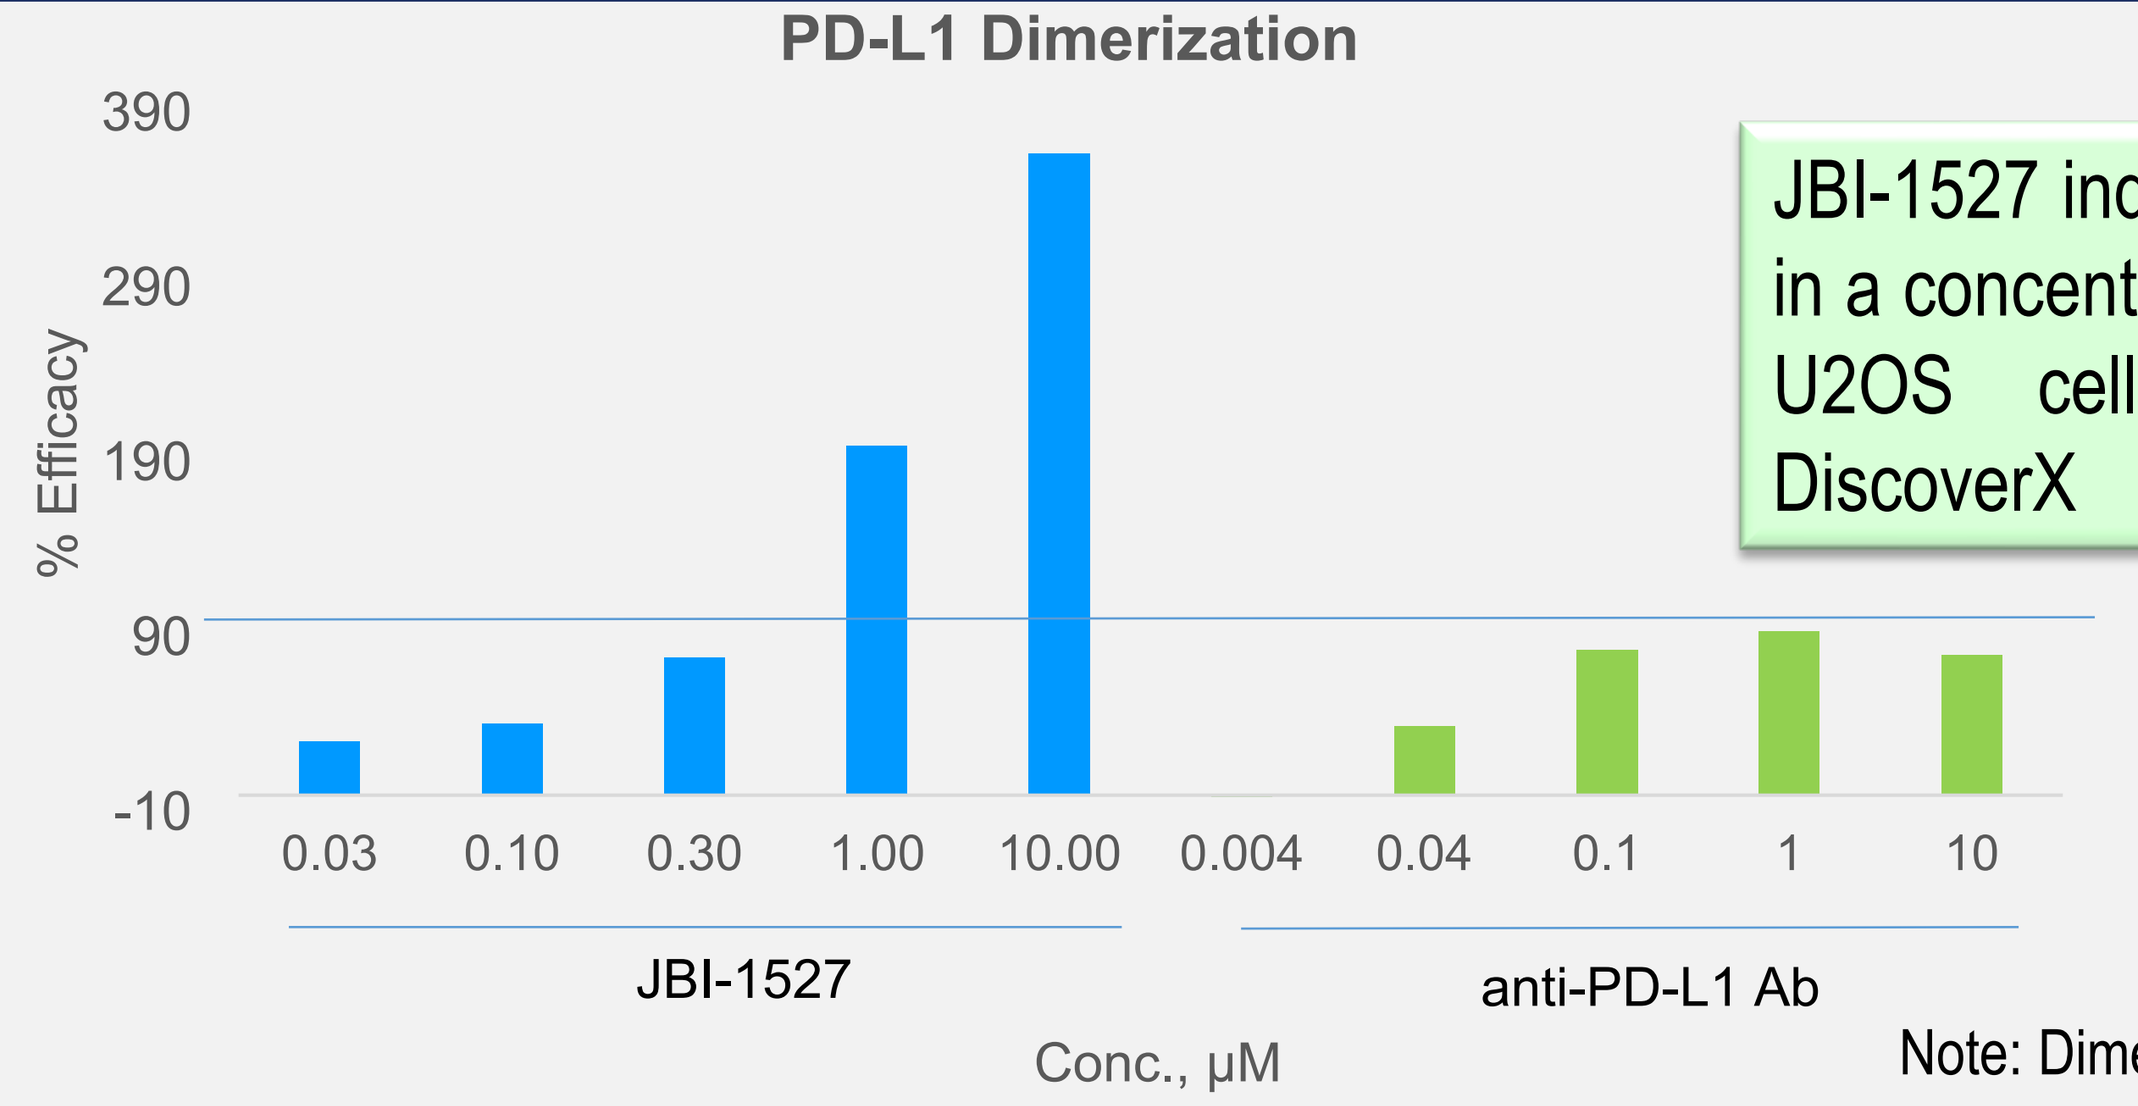
PD-L1 Dimerization
% Efficacy
JBI-1527
anti-PD-L1 Ab
Conc., μM

JBI-1527 induces dimerization of PD-L1 in a concentration dependent manner in U2OS cells (PathScan assay at DiscoverX

Note: Dimerization screening performed at DiscoverX

Co-crystal structure of JBI-1527: PD-L1 dimerization

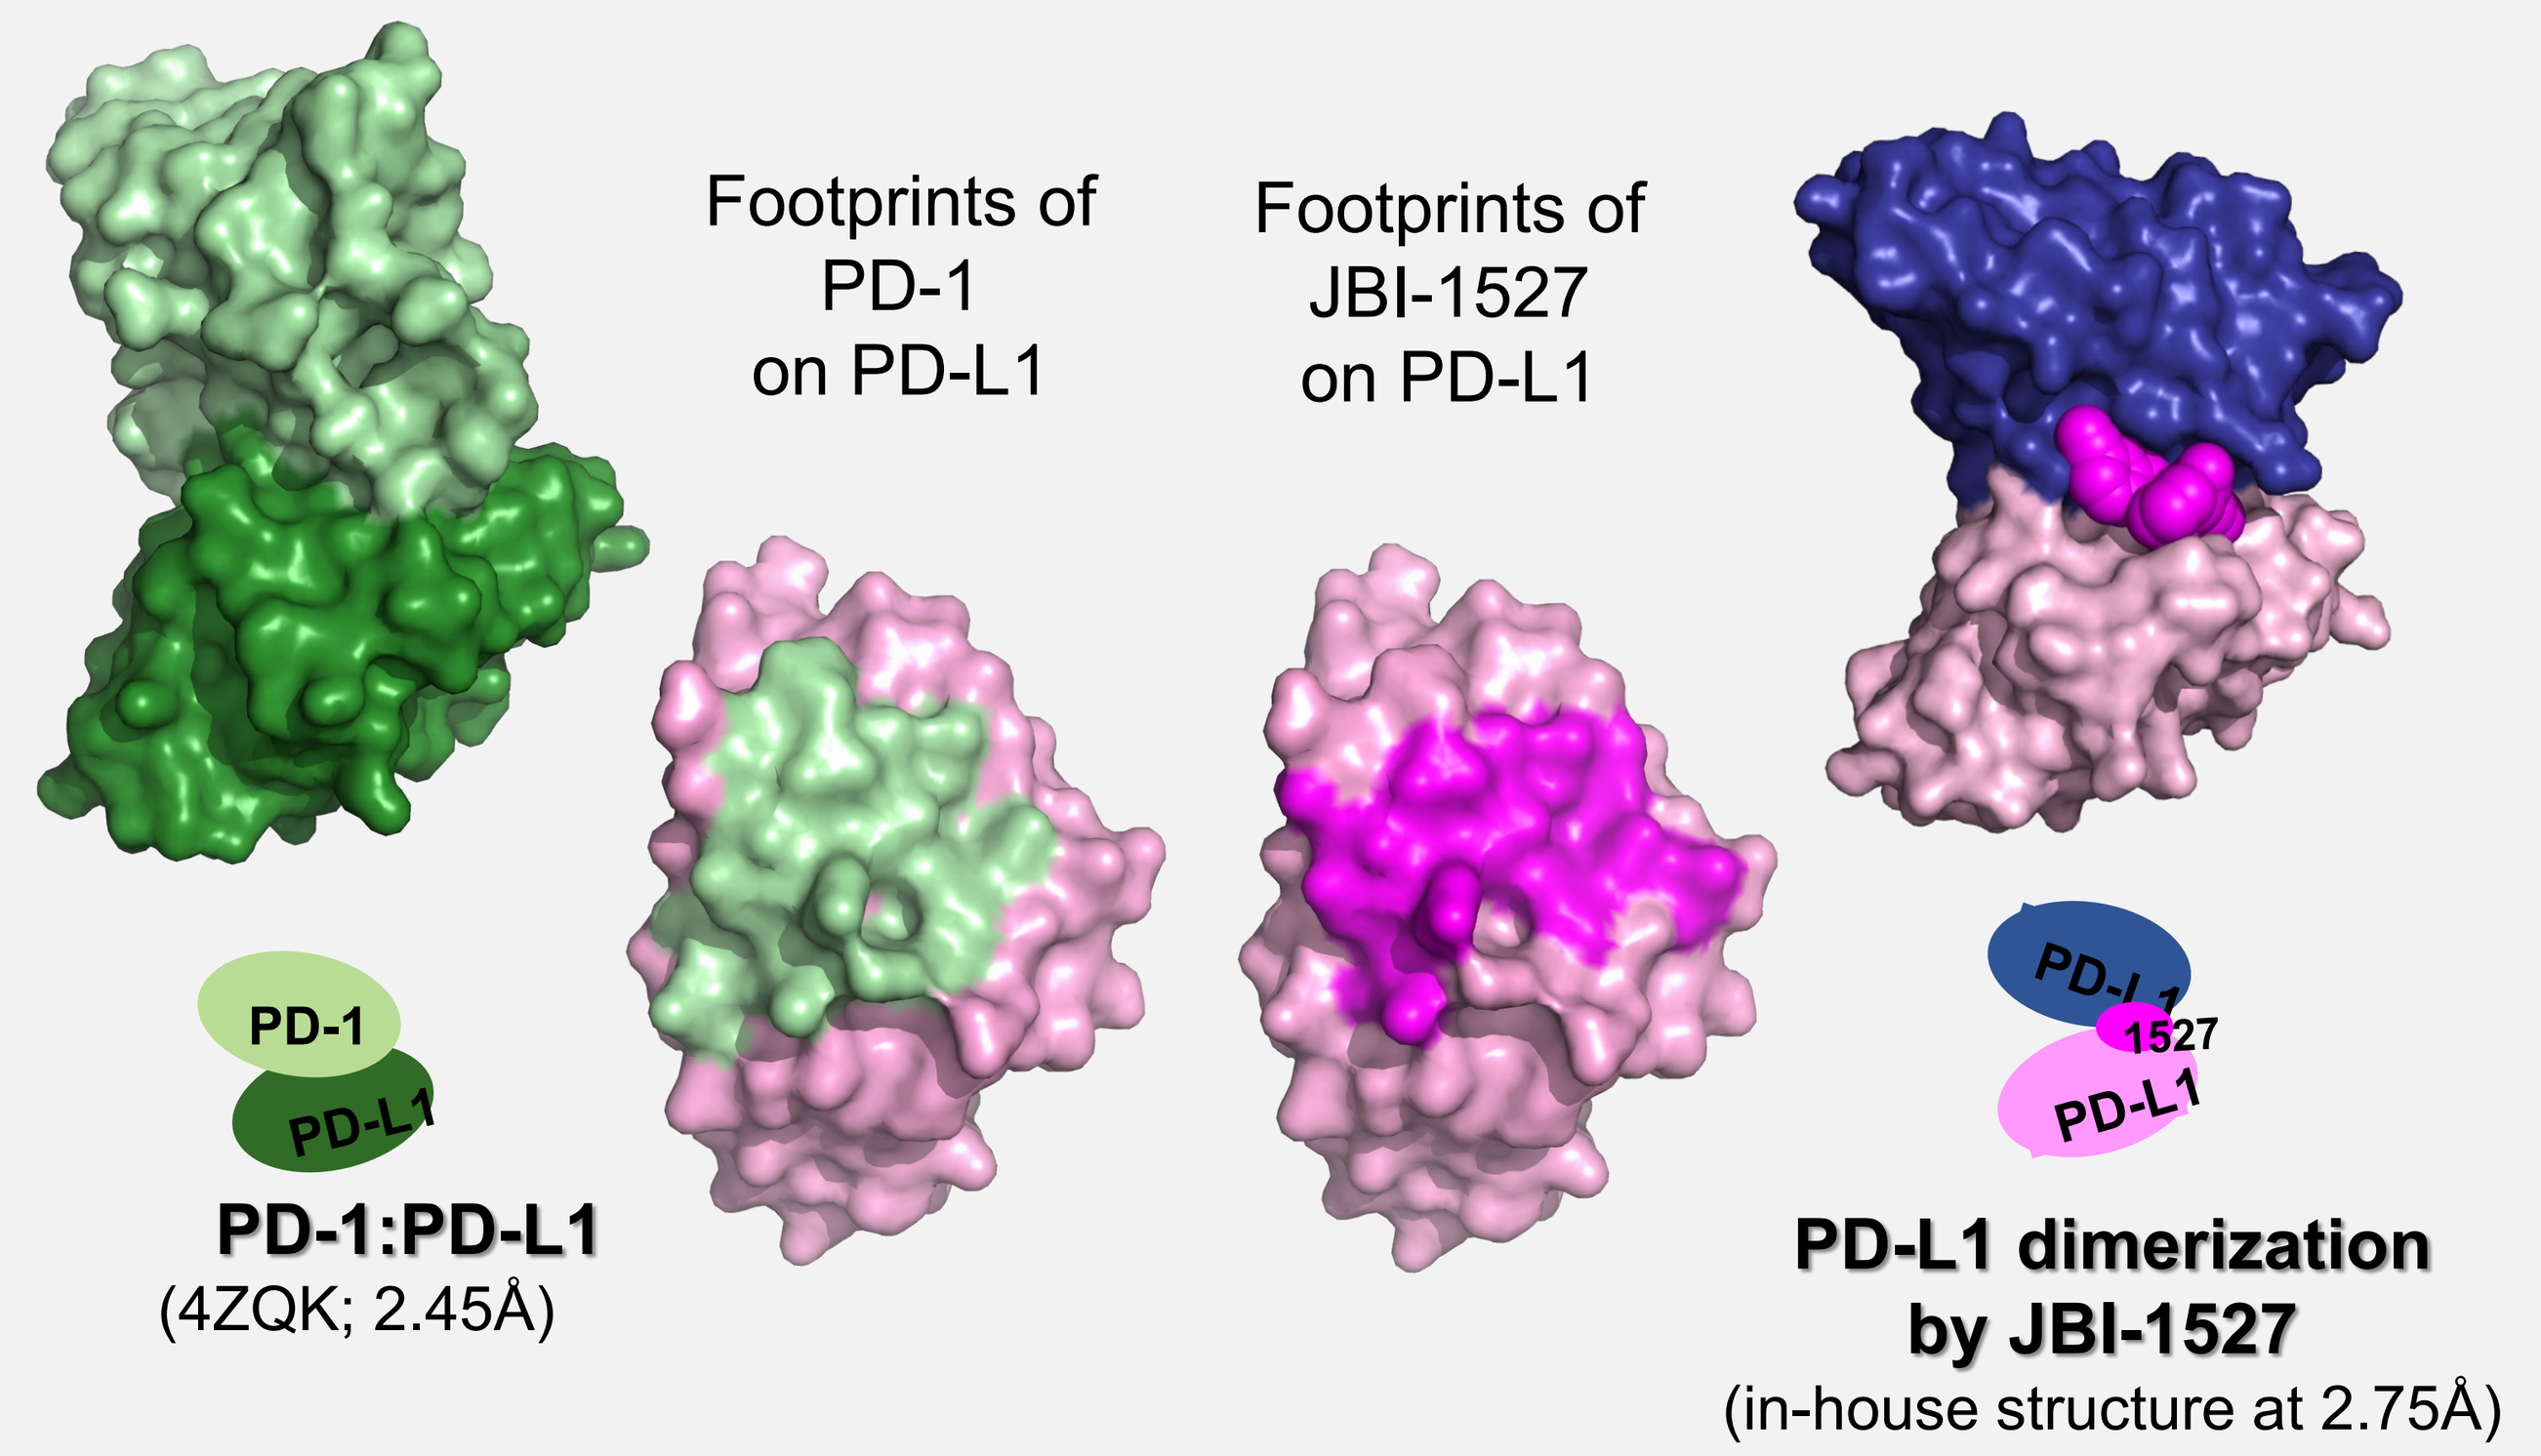
Footprints of PD-1 on PD-L1
Footprints of JBI-1527 on PD-L1
PD-1
PD-L1
PD-1:PD-L1
(4ZQK; 2.45Å)
1527
PD-L1 dimerization by JBI-1527
(in-house structure at 2.75Å)

JBI-1527 binds selectively to PD-L1 and inhibits interactions between PD1/PD-L1; leads to dimerization of PD-L1; JBI-1527 covers 58% of PD-1 footprint, similar to mAbs

JBI-1527 is efficacious in MC38-hPD-L1 model and induces TILs

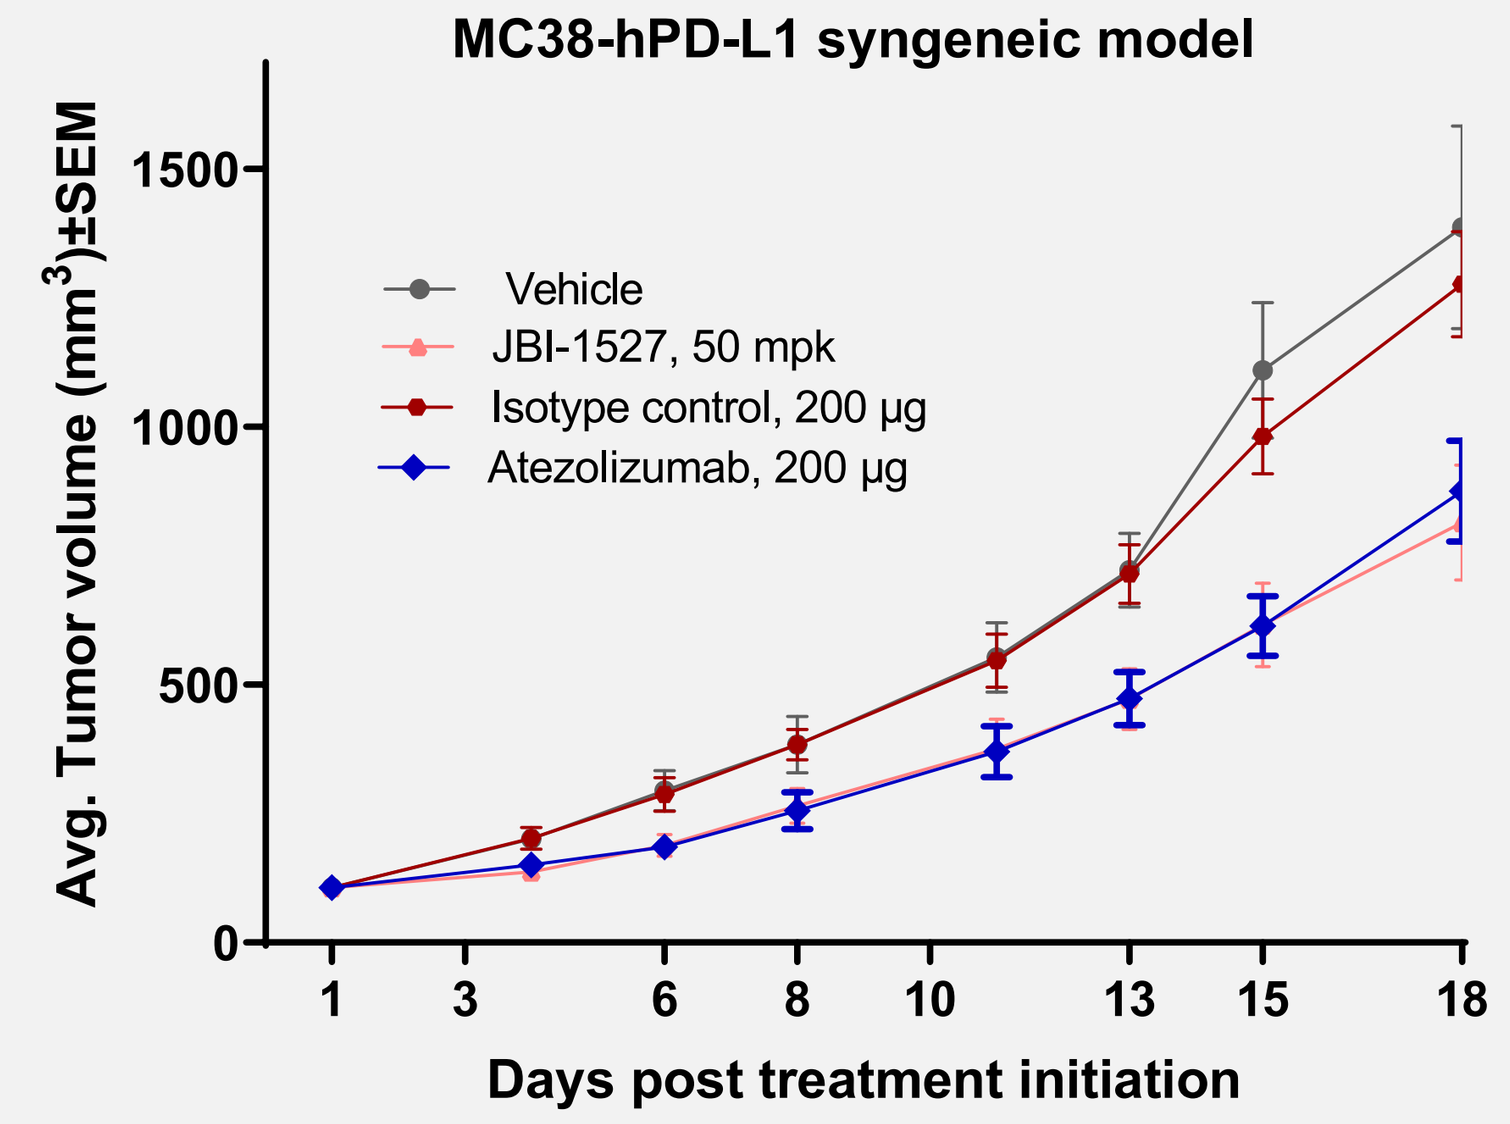
MC38-hPD-L1 syngeneic model
Avg. Tumor volume (mm3)±SEM
Vehicle
JBI-1527, 50 mpk
Isotype control, 200 μg
Atezolizumab, 200 μg
Days post treatment initiation

JBI-1527 showed strong tumor growth inhibition, comparable to Anti- PD-L1 mAb/Atezolizumab, and was well tolerated

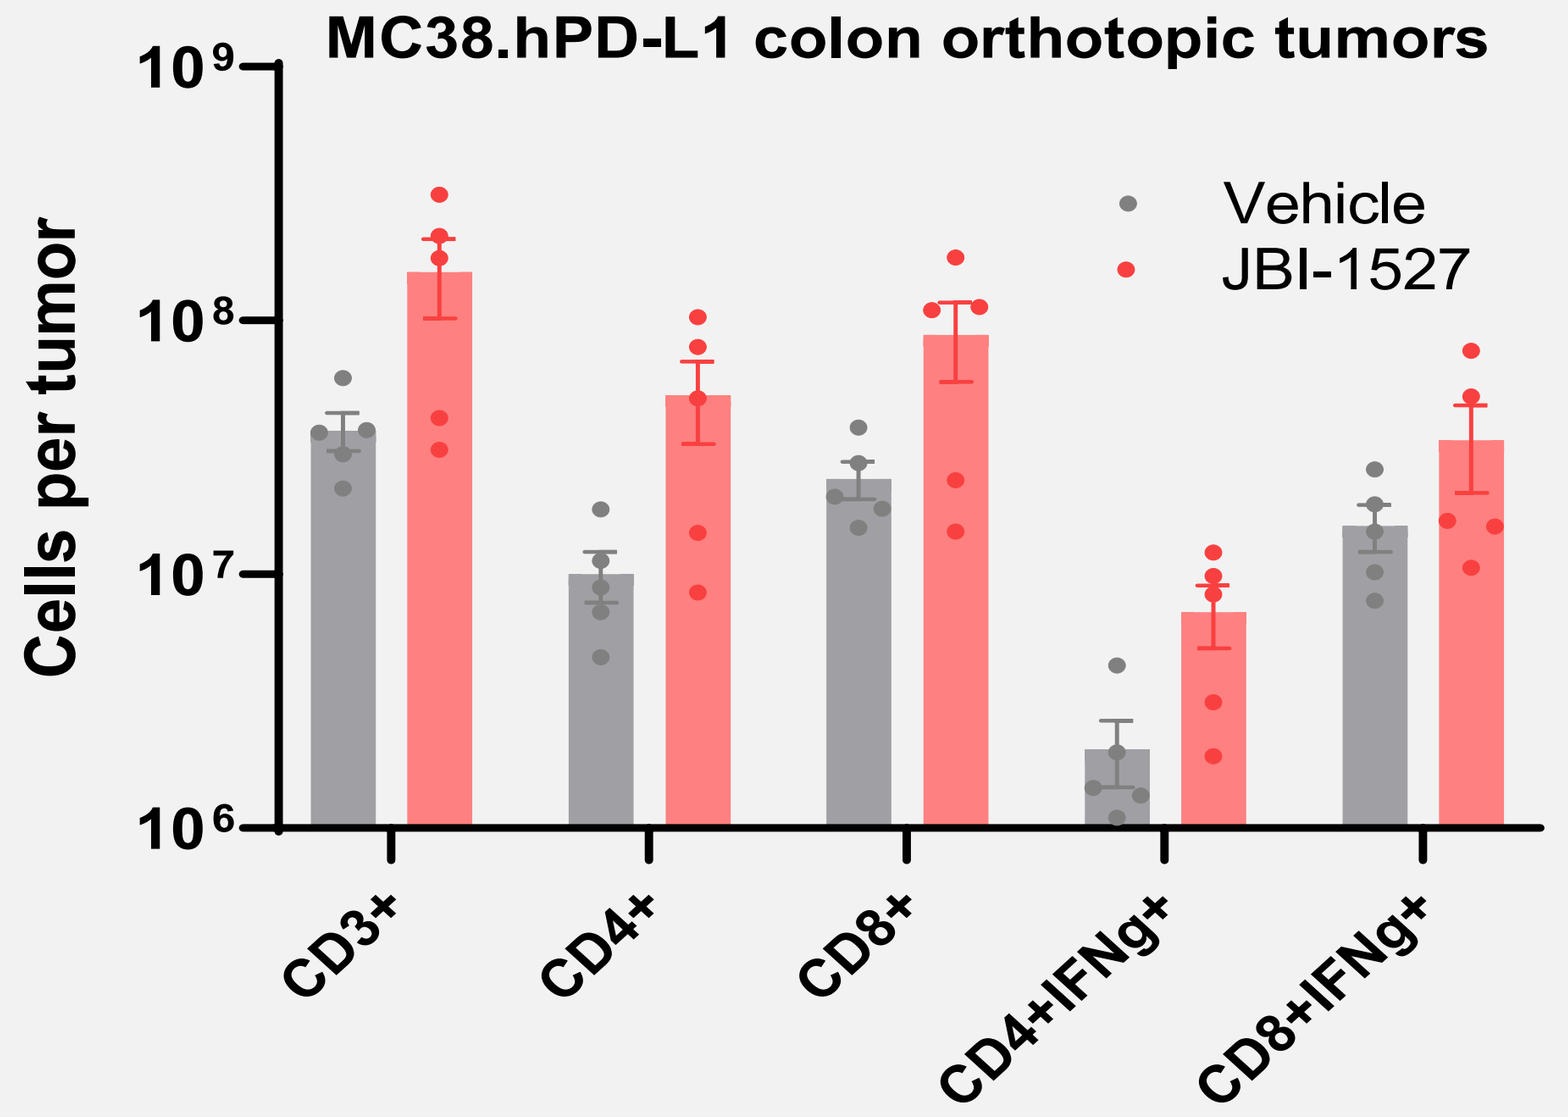
MC38.hPD-L1 colon orthotopic tumors
Cells per tumor
Vehicle
JBI-1527
CD3+
CD4+
CD8+
CD4+IFNg+
CD8+IFNg+

JBI-1527 induced infiltration of active T-cells in orthotopically implanted colon tumors

JBI-1527 shows a liver and gut-favorable exposure

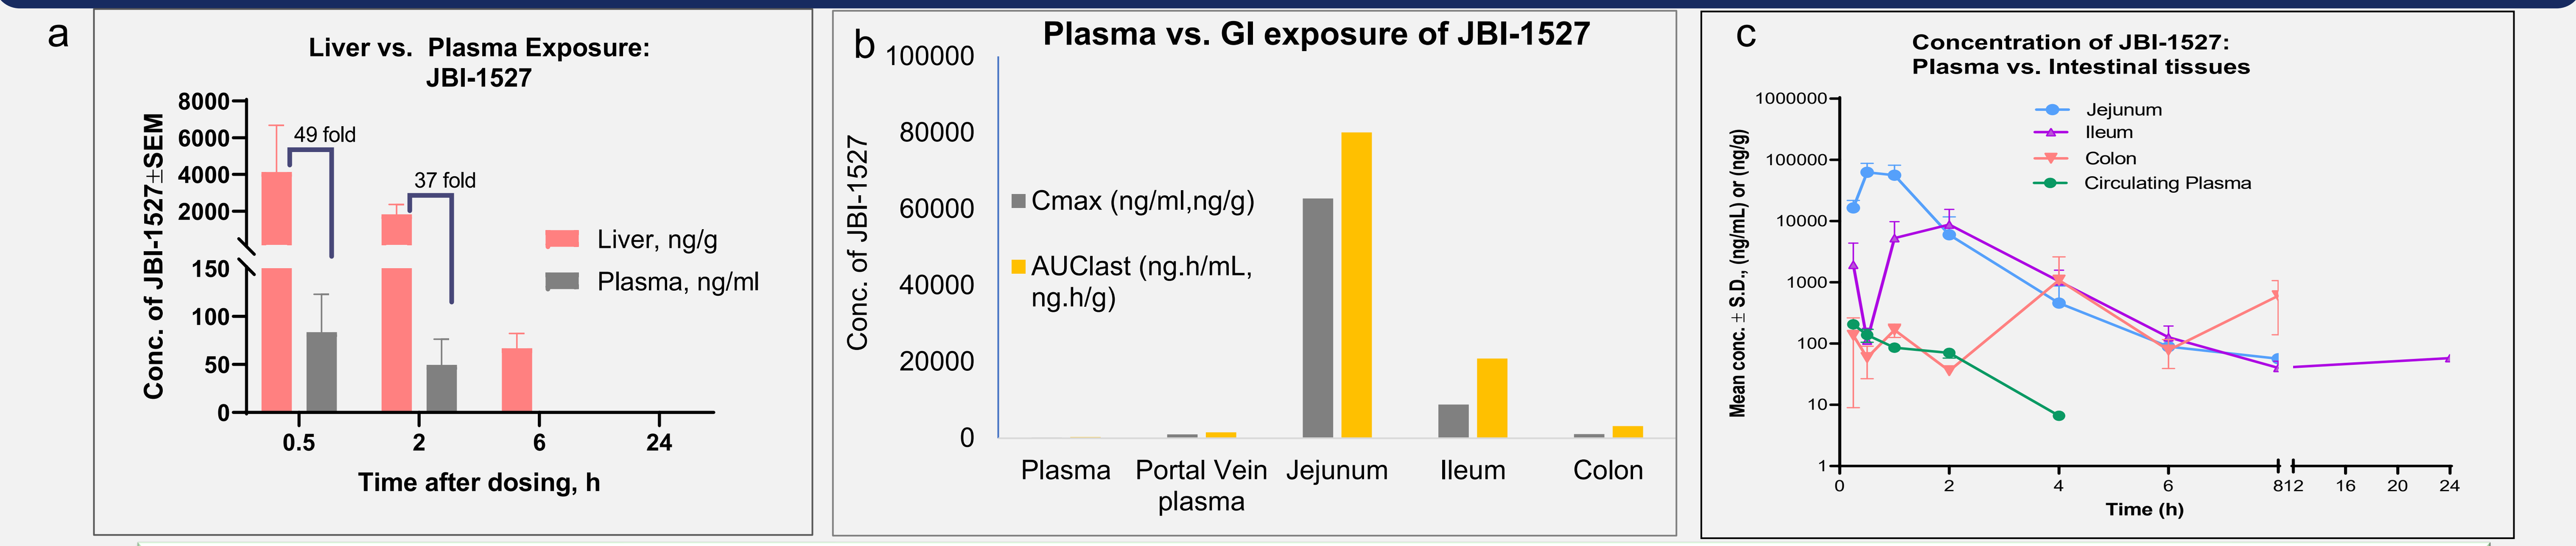
a
Liver vs. Plasma Exposure: JBI-1527
Conc. of JBI-1527±SEM
49 fold
37 fold
Liver, ng/g
Plasma, ng/ml
Time after dosing, h
b
Plasma vs. GI exposure of JBI-1527
Conc. of JBI-1527
Cmax (ng/ml,ng/g)
AUClast (ng.h/mL, ng.h/g)
Plasma
Portal Vein plasma
Jejunum
Ileum
Colon
c
Concentration of JBI-1527: Plasma vs. Intestinal tissues
Mean conc. ± S.D., (ng/mL) or (ng/g)
Jejunum
Ileum
Colon
Circulating Plasma
Time (h)

JBI-1527 shows much higher and sustained exposure in GI tract as compared to plasma a) Exposure of Liver vs Plasma b) Exposure of Plasma vs GI tract. c) concentration of Plasma vs Intestinal tissues

Conclusions

- Small molecule PD-1/PD-L1 inhibitors, can provide increased oral bioavailability, increased bio-efficiency and shorted half life activity for a more controllable treatment, in addition to having better safety specifically with regards to auto-immune related adverse effects, as compared to mAbs.
- Gastrointestine localised pharmacokinetic profile of JBI-1527 provides an attractive option to be used in the treatment of colon cancer, HCC and other GI-related cancers with minimal systemic toxicity as compared to mAbs
- Advanced toxicology studies are being initiated for this molecule

Acknowledgements

We would like to thank Amir Siddiqui M, Annasaheb S.kalange, Santosh Vishwakarma, Rudresh G and Sadanand Rangnathrao Mallurwar (Jubilant Biosys Ltd, Bangalore, India) for assistance in conducting the experiments.

Contact: luca.rastelli@jubilanttx.com / Dhanalakshmi.Sivanandhan@jubilanttx.com

Do Not Post